A randomized, placebo-controlled trial to evaluate the efficacy and safety of BIO101 on Sarcopenic Obesity.

Waly Dioh[1], Philippe Guillet[1], Susanna Del Signore[1,2], René Lafont[1,3] and Stanislas Veillet[1]

1. Biophytis. Parc BIOCITECH, 102 avenue Gaston Roussel. 93230 Romainville France; 2. Bluecompanion LTD, London, UK; 3. Sorbonne Universités, Université Pierre et Marie Curie, IBPS, 7 quai Saint Bernard, 75005 Paris. France.

INTRODUCTION

Sarcopenic obesity (SO), an emerging condition defined by the loss of muscle mass and function and a concomitant increase of fat mass, is mainly affecting older obese individuals. Establishing a consensus definition of SO has represented the first challenge, since most of previous studies specifically focused on body composition (gain of fat mass and loss of muscle mass) and not on physical function. SO definition also reveals the problem of its wide range prevalence (4-84% in men and 4-94% in women depending on definitions and body composition methods used; Batsis et al., 2013) and consequently of its recognition as a geriatric condition. Thus, despite SO high relevance, appropriate clinical studies associating body composition and physical function measurements are needed to better define its prevalence in the ageing population.

We developed the drug candidate Active Pharmaceutical Ingredient (API) BIO101, by purifying 20-hydroxyecdysone (>97%) from *Stemmacantha carthamoides.* Sarconeos is the investigational pharmaceutical product formulated with BIO101 for the treatment of sarcopenia including sarcopenic obesity. 20-hydroxyecdysone (20E), a polyhydroxylated sterol found in plants and pharmacologically active in mammals, has been reported *in vitro* to inhibit myostatin gene expression and to enlarge muscle fibers. These effects involve the activation of MAS receptor, a GPCR, whose endogenous ligand is Angiotensin 1-7. *In vivo*, 20E oral treatment prevents adipose tissue development, decreases myostatin gene expression in muscles, while enhancing expression of several markers of myogenesis. In the few clinical trials reported up to now, 20E induced fat mass reduction in obese subjects, and reduced lean mass loss in athletes submitted to intense physical training.

We aim to perform a Phase 2 trial on sarcopenic including sarcopenic obese elders. Given the unknown prevalence of sarcopenic obesity in the pre-identified clinical investigation centers, we will first perform pilot studies in Europe and USA to better characterize the target population and to facilitate subsequent enrollment in the main clinical trial (CT). We are also performing safety and pharmacokinetic studies in order to confirm previous results obtained with 20E. The design of both preparatory studies are presented here and will clearly pave the way towards the Phase 2 clinical trial with BIO101.

Prevalence of Sarcopenic Obesity depending on definitions and approaches

References	Sample size and age	Obesity		Sarcopenia		Sarcopenic Obesity	
		Definition	Prevalence in %	Definition	Prevalence in %	Definition	Prevalence in %
Kemmler et al., 2016	1325 Community Dwelling women ≥70 years	Body Fat mass ≥ 35%	68	EWGSOP	4.5	EWGSOP + BMI ≥ 30 kg/m²	0%
		BMI ≥ 30 kg/m²	19.8	IWGS	3.3	EWGSOP + Body Fat mass ≥ 35%	2.3%
Batsis et al., 2014	4652 subjects ≥ 60 years	Body Fat Mass ≥ 27% in Men.	54.4%	Total SM/ht²⩽ 5.75 kg/m² in Men.	75.5%	Body Fat Mass ≥ 27%, and Total SM/ht² ⩽ 5.75 kg/m2 in Men.	18.1 %
		Body Fat Mass ≥ 38% in Women	60.8%	Total SM/h²⩽10.75 kg/m² in Women	35.4%	Body Fat Mass ≥ 38% and Total SM/h²⩽10.75 kg/m2 in Women	42.9 %
Batsis et al., 2015	4984 subjects ≥60 years : 2453 men and 2531 women	Body Fat Mass ≥25% in Men	NA	ALM <19.75 kg,	16%	ALM or ALM/BMI + Body Fat mass % ≥25%, in Men	12.5% for ALM
				ALM/BMI <0.789 kg/m².	27.8%		27.3% for ALM/BMI
		Body Fat Mass ≥ 35% in Women	NA	ALM <15.02 kg	40.5%	ALM or ALM/BMI + Body Fat mass % ≥35%, in Women	33.5% for ALM
				ALM/ BMI <0.512 kg/m2	19.3%		19.1% for ALM/BMI
Bouchard et al., 2009	904 subjects: 432 men and 462 women 60-82 years	Body Fat Mass ≥ 28% in Men	49%	ASMI < 8.51 kg/m² in Men	38%	Body Fat Mass + ASMI in Men	18%
		Body Fat Mass ≥ 35% in Women	70%	ASMI < 6.29 kg/m2 in Women	18%	Body Fat Mass + ASMI in Women	11%
Dufour et al., 2012 Limitation in at least 1 mobility item : 30% of women; 16% of men	767 subjects: 274 Men and 493 women. 72-92 years	Body Fat Mass ≥ 30% in Men	19%	ALM/ht ² : < 7.26 kg/m2 in men	NA	ALM/ht2 + Body Fat Mass > 30% in Men	8%
		Body Fat Mass ≥ 40% in Women	13%	and < 5.45 kg/m² in women	NA	ALM/ht 2 : < 5.45 kg/m2 + Body Fat Mass > 40% in Women	4%
Rolland et al., 2009	1308 healthy women aged > 75 years	Body Fat mass > 40%	33.2%	Appendicular skeletal muscle mass (ASM) < 5.45 kg/m²	6.8%	ASM<5.45 kg/m2	2.75%
Yang et al., 2015	844 subjects : 448 men and 396 women > 65 years.	Body Fat Mass > 27.82% in men		SMI < 6.87 kg/m² in men		Body Fat Mass > 27.82 % and SMI < 6.87 kg/m² in Men	7.37%
		Body Fat Mass > 37.61% in women	32.94%	SMI < 5.46 kg/m² in women,	11.85%	Body Fat Mass > 37.86 % and SMI < 5.46 kg/m² in Women.	7.07%

The selected studies show wide range of prevalence for sarcopenic obesity. This recommends to perform pilot studies before initiating the Sarconeos Phase 2 clinical trial. Pilot studies will be held in Europe and USA.

SARA-obs: Characterisation of Sarconeos Phase 2 target population in Europe and USA

Subjects aged ≥ 65 years complaining for Poor Physical function

SPPB Measurement

Score < 8/12

Grip Strength

Strength < 26 kg in Men and < 16 kg in Women

DEXA Scan

ALM/BMI < 0.789 in men and 0.512 in women And Body fat Mass > 28% in men and 35% in women

- **6 minutes distance**
- **400 meters walk**

The pilot study will provide key information on the applicability of selection/inclusion criteria for the phase 2 interventional trial.

SARA-obs clinical study design

Goals :

- **Characterize the target population for the Phase 2 in Europe and USA.**
- **Estimate the prevalence of sarcopenia including sarcopenic obesity in a representative sample of older persons with poor physical function.**
- **Measure the variability of the tentative main criteria: 6- minute walk distance or 400-m walking tests.**

Design :

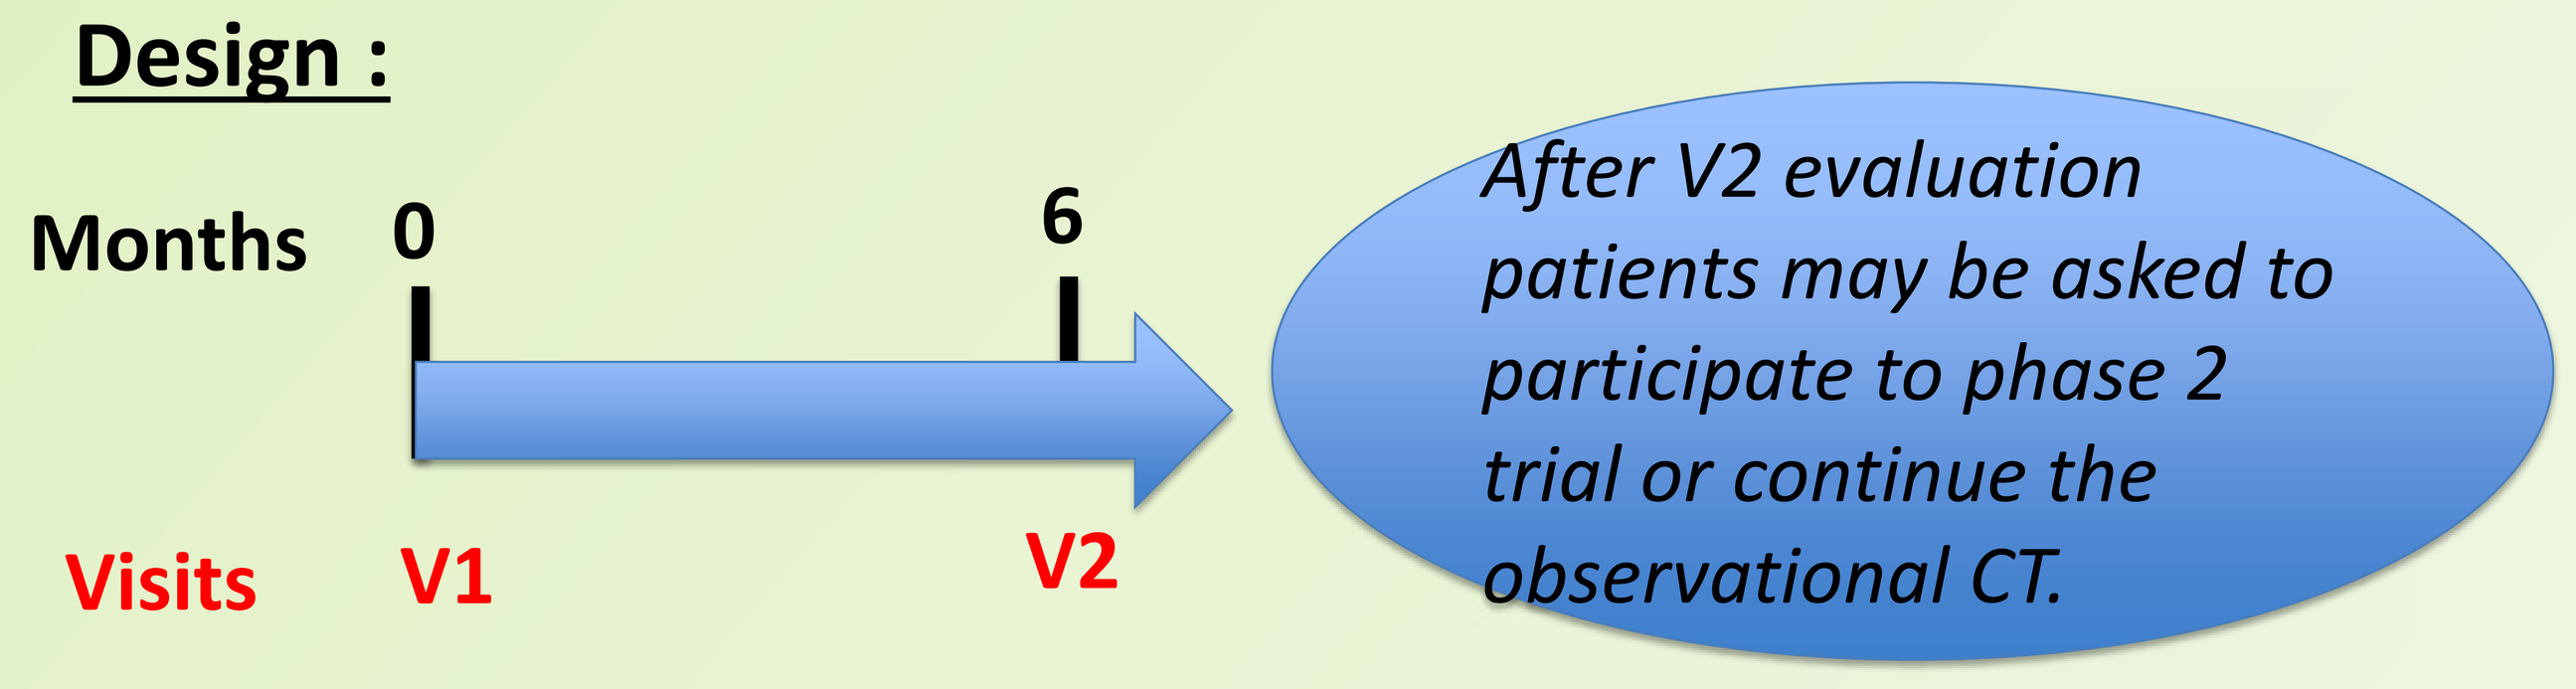
Months
0
6
Visits
V1
V2
After V2 evaluation patients may be asked to participate to phase 2 trial or continue the observational CT.

Parameters :

Main endpoints: **6 minutes walk/ 400 m walking test**

Secondary endpoints: **Body composition**

Gait speed; Grip strength

The pilot study design consisted of two main visits (once every 6 months) to evaluate the main endpoints, including 6-minute walk and 400-meter, and the secondary endpoints including body composition, gait speed and grip strength.

The retained subjects will be asked to participate in the subsequent Sarconeos interventional phase 2 clinical trial.

SARA-pk: Safety and pharmacokinetics of Sarconeos in healthy young and older adults volunteers after single ascending oral doses

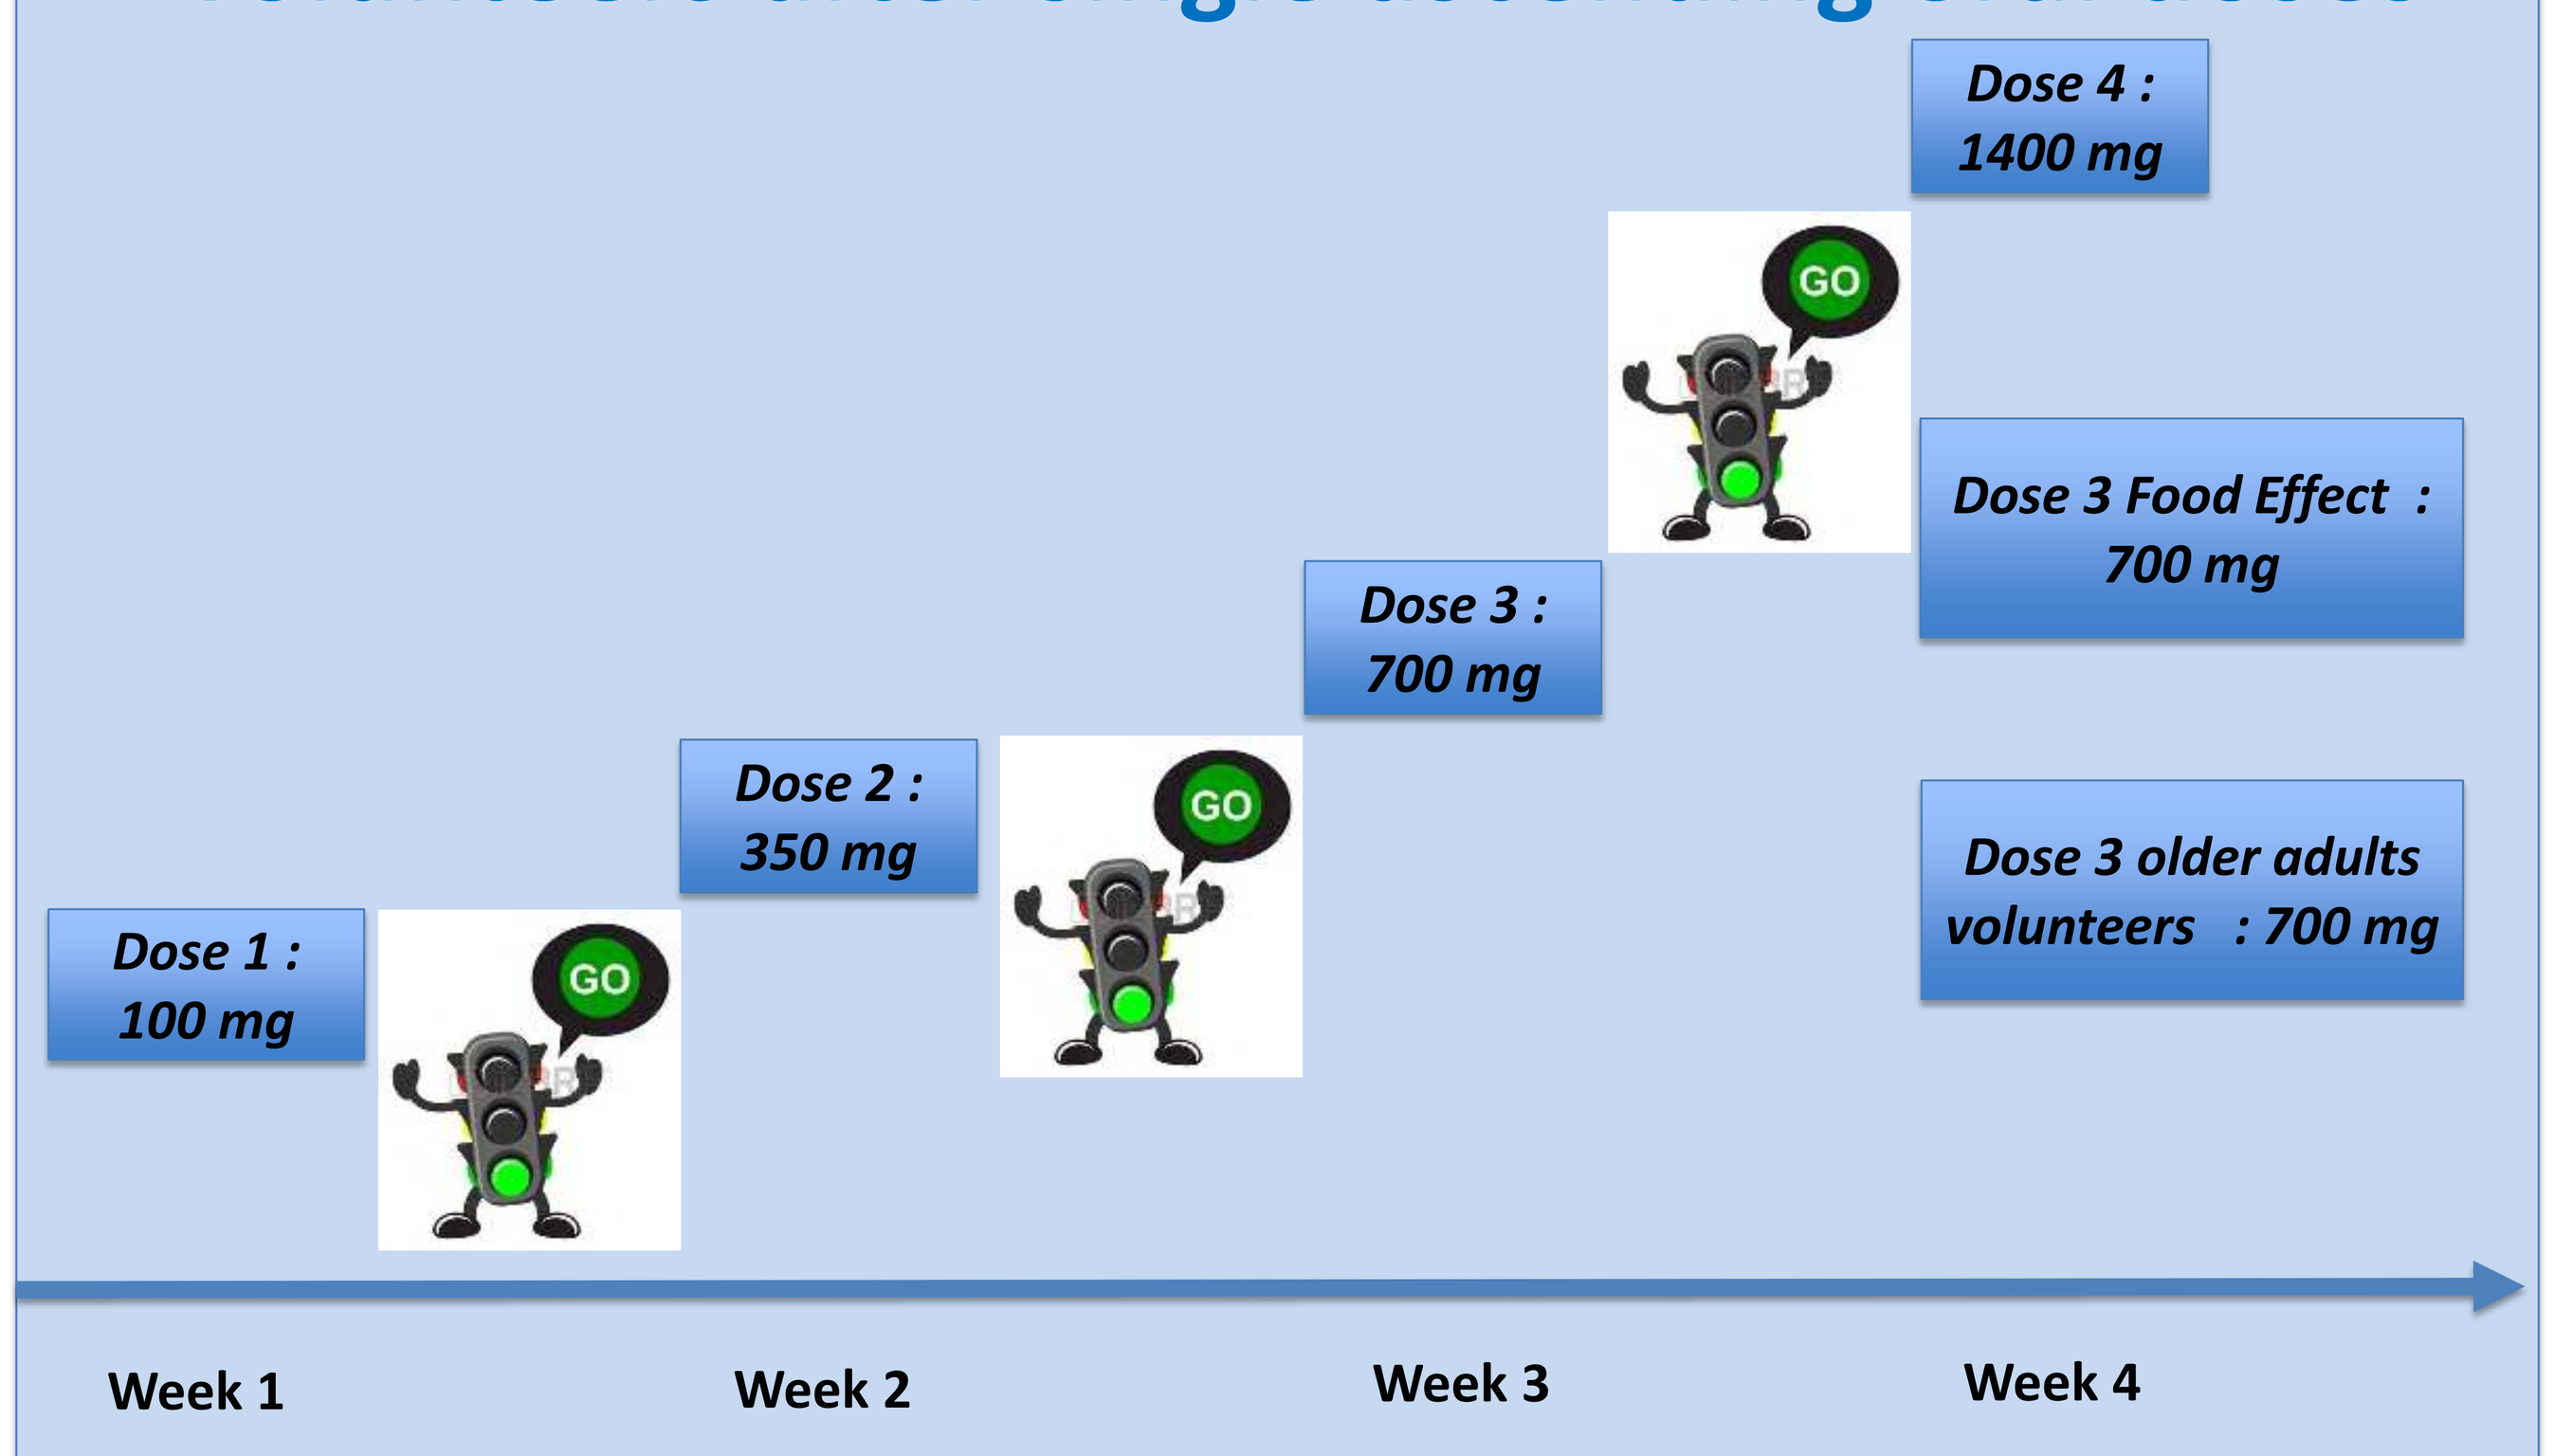
Dose 1 : 100 mg
Dose 2 : 350 mg
Dose 3 : 700 mg
Dose 4 : 1400 mg
Dose 3 Food Effect : 700 mg
Dose 3 older adults volunteers : 700 mg
GO
Week 1
Week 2
Week 3
Week 4

- **Safety and Pharmacokinetics of Sarconeos will be evaluated in a single ascending dose study.**
- **The food effect will be also evaluated on Sarconeos pharmacokinetics.**
- **Pharmacokinetics will be compared between young and older adults.**

SARA-pk: Safety and pharmacokinetics of Sarconeos in healthy older adults volunteers after multiple ascending oral doses for 14 days.

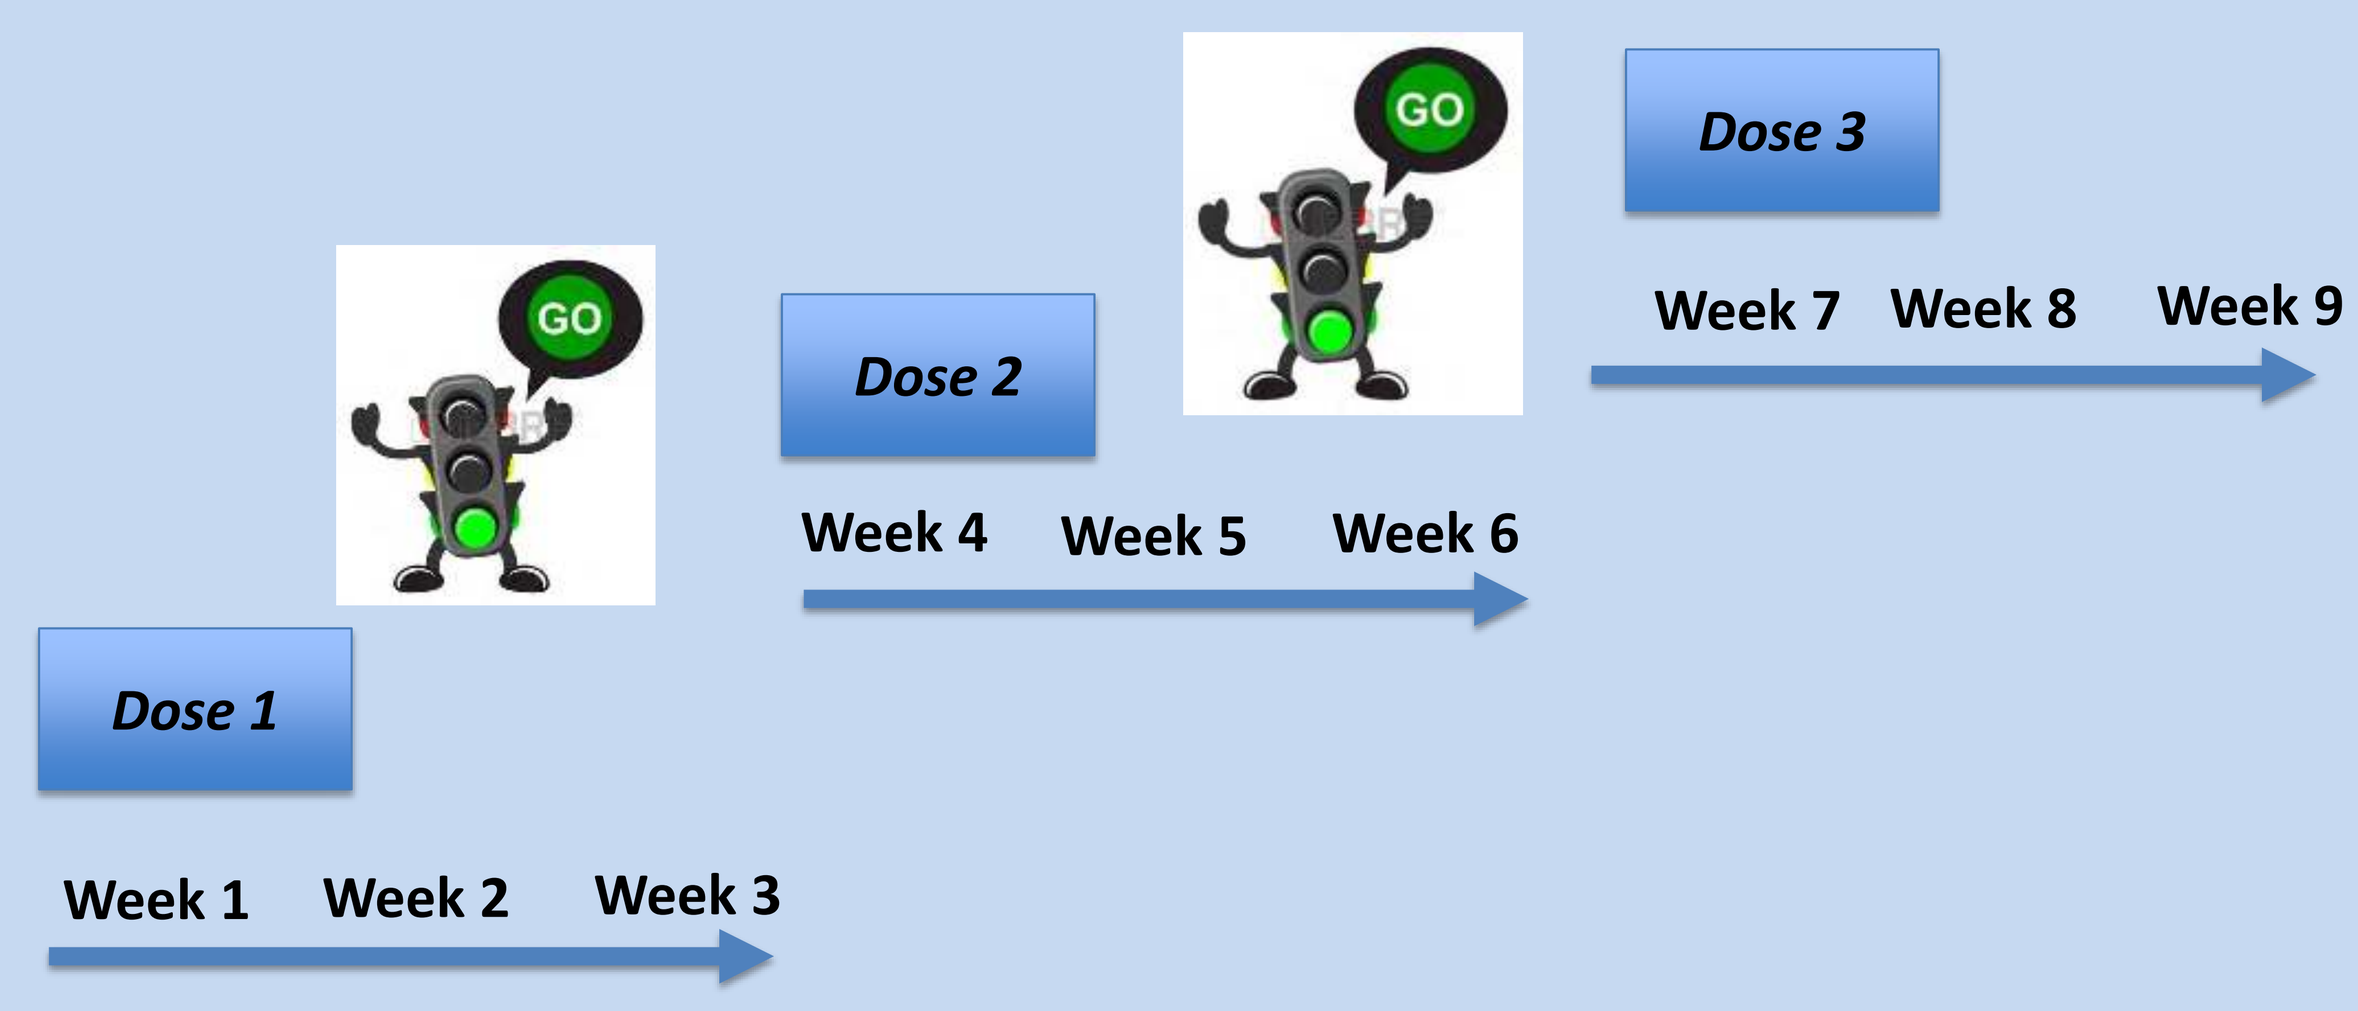
Dose 1
Week 1
Week 2
Week 3
Dose 2
Week 4
Week 5
Week 6
Dose 3
Week 7
Week 8
Week 9
GO

- **Safety and Pharmacokinetics of Sarconeos and its main metabolites will be evaluated in a multiple ascending dose during 14 days.**
- **Plasmatic and urinary pharmacodynamic markers will be assayed :**

1. **Markers of skeletal muscle proteolysis and proteosynthesis**
2. **Markers of the Renin Angiotensin Aldosterone System.**

CONCLUSIONS

Sarconeos Efficacy and Safety CT in Sarcopenic Obesity of older Patients (SARA-int) will be implemented both in the EU and USA.

Prior to the main study, a pilot observational CT (SARA-obs) will allow to characterize sarcopenic obesity subpopulation in the framework of sarcopenia, based on the wide range of reported prevalence of this condition.

Additionally, a PK/PD CT (SARA-pk) will be conducted in older adults.

These preparatory studies will pave the way for a smoother implementation of SARA-int, the planned interventional Phase 2 CT.