BIO103 a drug candidate for the treatment of muscle wasting disorders

Maria Serova[1], Blaise Didry-Barca[1], Sissi On[1], Anne-Sophie Foucault[1], Sophie Raynal[1], Stanislas Veillet[1], Pierre Dilda[1], René Lafont[2]

[1] Biophytis, UMPC – BC9, 4 place Jussieu, 75005 Paris, France
[2] Sorbonne Universités, UPMC Univ Paris 06, CNRS - Institut de Biologie Paris Seine (BIOSIPE), 75005 Paris, France

Introduction

Muscle wasting disorders, including cachexia and sarcopenia, are multifactorial diseases which contribute to overall physical frailty. They represent a worldwide health challenge with limited therapeutic options. Many cellular factors were identified to maintain normal muscle function. Cell metabolism influenced by AKT/mTOR and mitochondrial biogenesis in parallel with physical activity contribute to maintaining of muscle mass and strength. Age-related deregulation of these mechanisms leads to muscle wasting.

About ecdysteroids: The steroid hormone (20E) plays a key role in insect development through nuclear ecdysone receptors (EcRs) and at least one membrane receptor (DopEcR). 20E also displays pharmacological effects on mammals, where it stimulates protein synthesis although EcR were not found in mammals.

About BIO103: BIO103, ecdysteroid derivative, is the product of hemisynthesis associated with the screening of more than 100 derived molecules. This screening had the object of selecting a molecule with improved activity and bioavailability.

The aim of this study was to characterize a new small molecule, BIO103, *in vitro* on myocytes and *in vivo* on a mouse model designed to analyze the effects of aging and muscle disuse.

Proposed mechanism of action

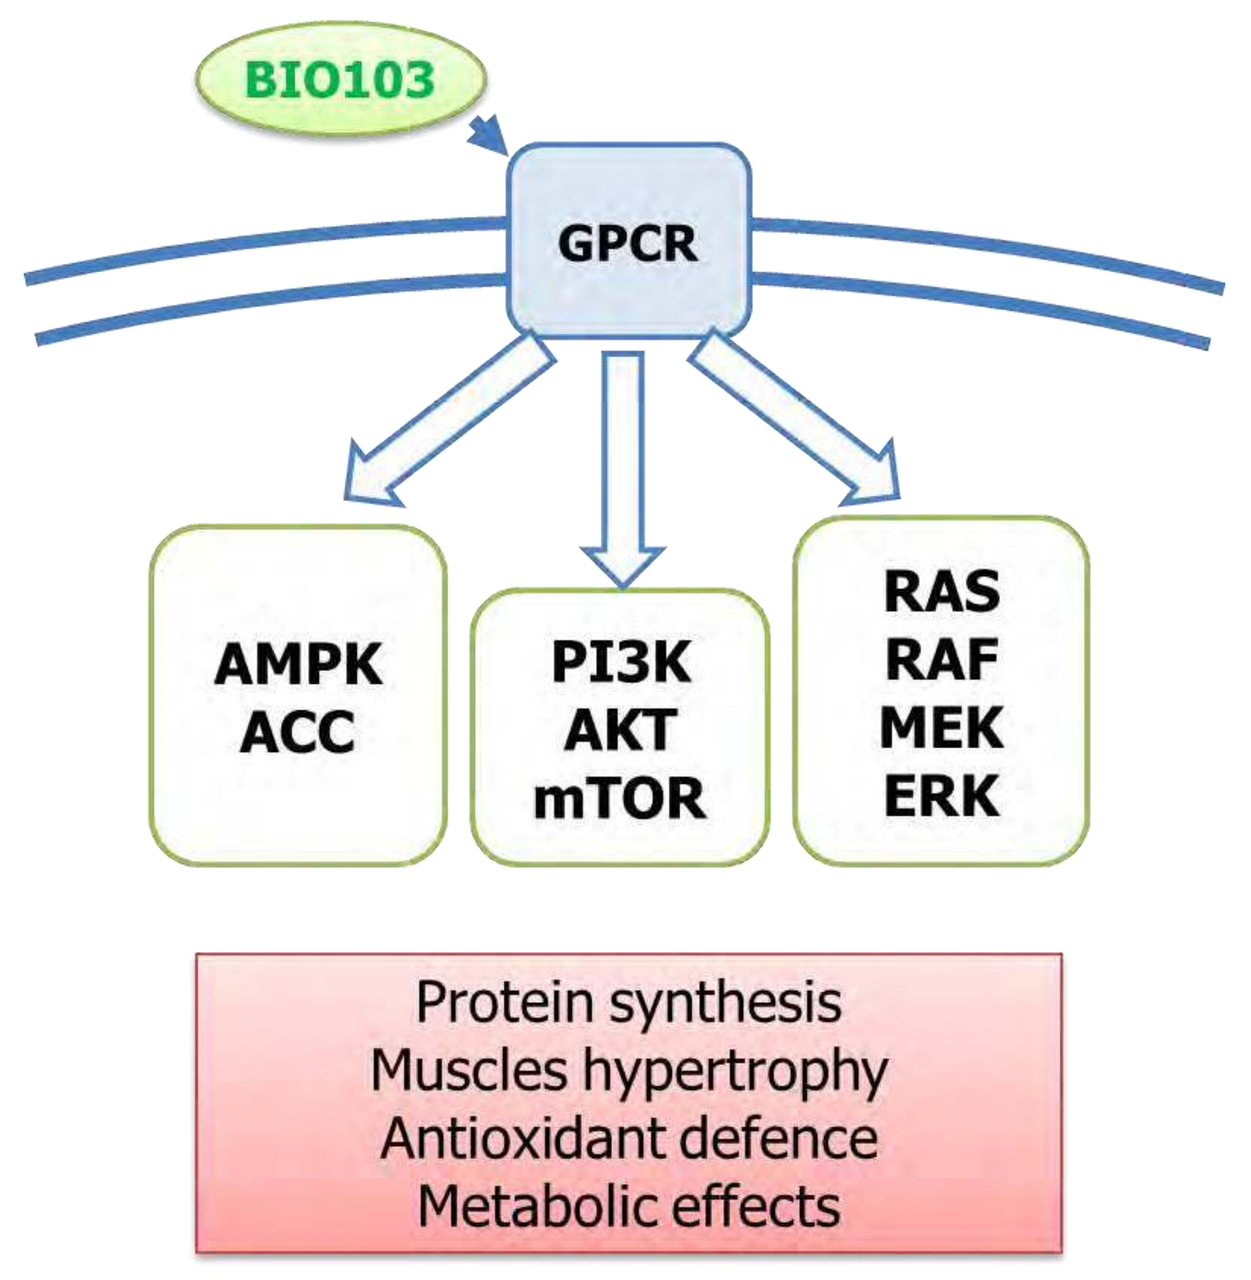

BIO103
GPCR
AMPK ACC
PI3K AKT mTOR
RAS RAF MEK ERK
Protein synthesis
Muscles hypertrophy
Antioxidant defence
Metabolic effects

Methods

Cell line: C2C12 murine myoblasts were induced for differentiation for 5 days. Appropriate treatment was administered for 6h (for gene expression analysis) or 4 days (for fluorescent microscopy).

Gene expression: Total mRNA was extracted and purified using Trizol method. mRNAs were reverse-transcribed into cDNAs and Myostatin gene expression was analyzed by quantitative RT-PCR. HPRT was used as housekeeping gene.

Immunofluorescence: Cells were grown, differentiated and treated on 8 well chamber slides. Then the cells were fixed with glutaraldehyde 2.5%/triton 0.1%, covered by DAPI-containing mounting medium. After 24h in the dark, myotubes were observed under fluorescent microscope.

Western blot: Cells were lysed, equal amounts of proteins were electrophoresed on 4-12% SDS-PAGE and transferred to nitrocellulose membranes. Membranes were blocked with 5% non-fat milk and incubated with specific antibodies overnight. Immunostaining was visualized using ECL. Bands intensity was quantified using ImageJ software.

OCR measurements: Oxygen consumption was recorded using a Seahorse XF Analyzer.

In vivo studies. Adult (12 months) and old (22 months) C57Bl6/J female mice were used. The old animals were treated orally for 14 weeks with either vehicle or BIO103 (50 mg/kg). After 13 weeks of treatment), the animals were tested for functional activity in toto (Vmax). Plasma and muscles were collected after sacrifice for further analysis

Results

- **In vitro**

Effects of BIO103 on C2C12 myotube diameter

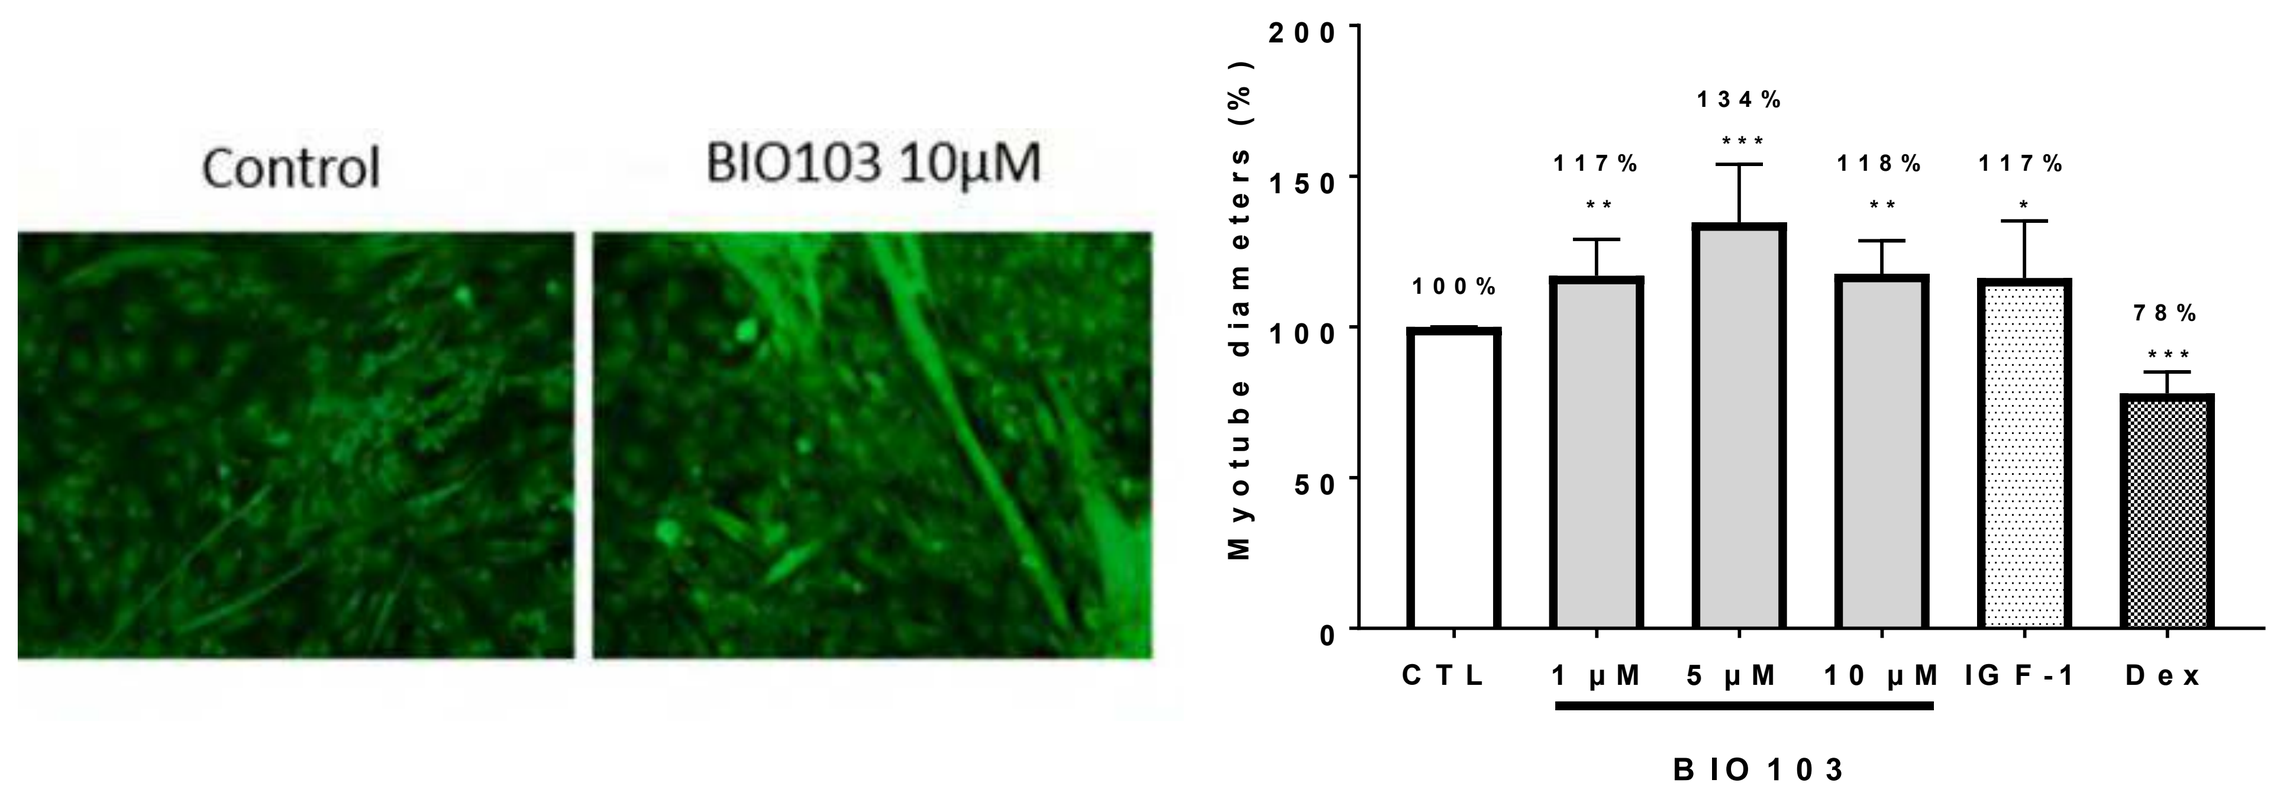

Control
BIO103 10µM
Myotube diameters (%)
100%
117%
134%
118%
117%
78%
CTL
1 µM
5 µM
10 µM
IGF-1
Dex
BIO103

→ 3-day treatment of differentiated C2C12 myocytes with BIO103 (1, 5, 10 µM), induced significant increase of myotube diameters

Effect of BIO103 on myostatin gene expression

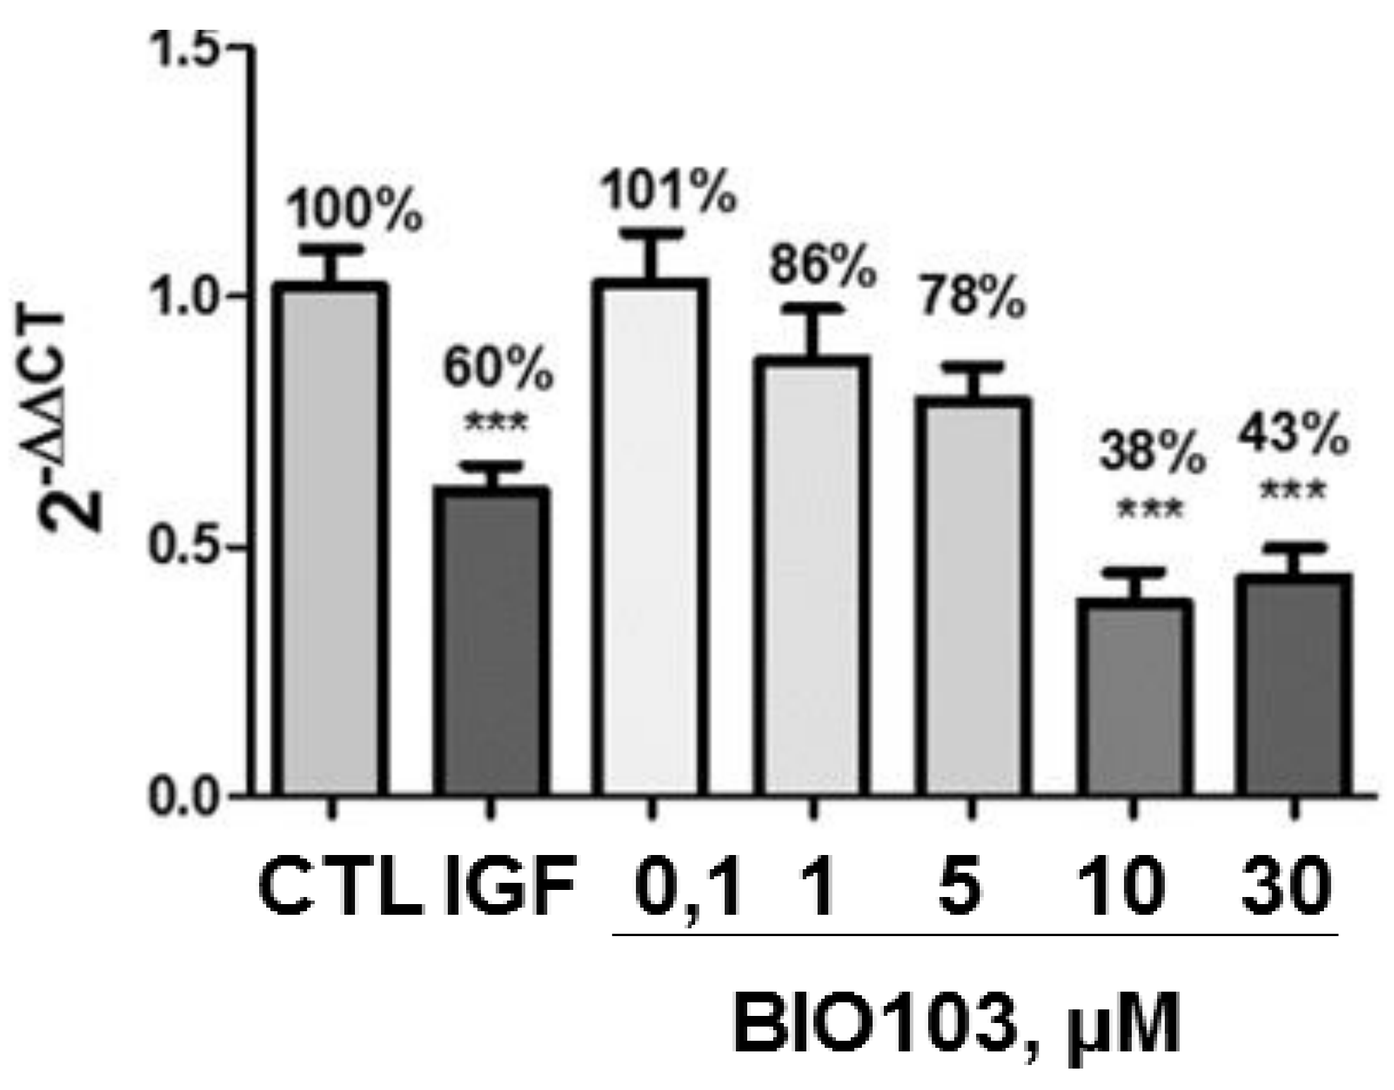

2-ΔΔCT
100%
60%
101%
86%
78%
38%
43%
CTL
IGF
0,1
1
5
10
30
BIO103, µM

→ BIO103 6 hour-treatment of differentiated C2C12 cells significantly inhibits myostatin gene expression

Effects of BIO103 on intracellular signaling

- PI3K/AKT/mTOR

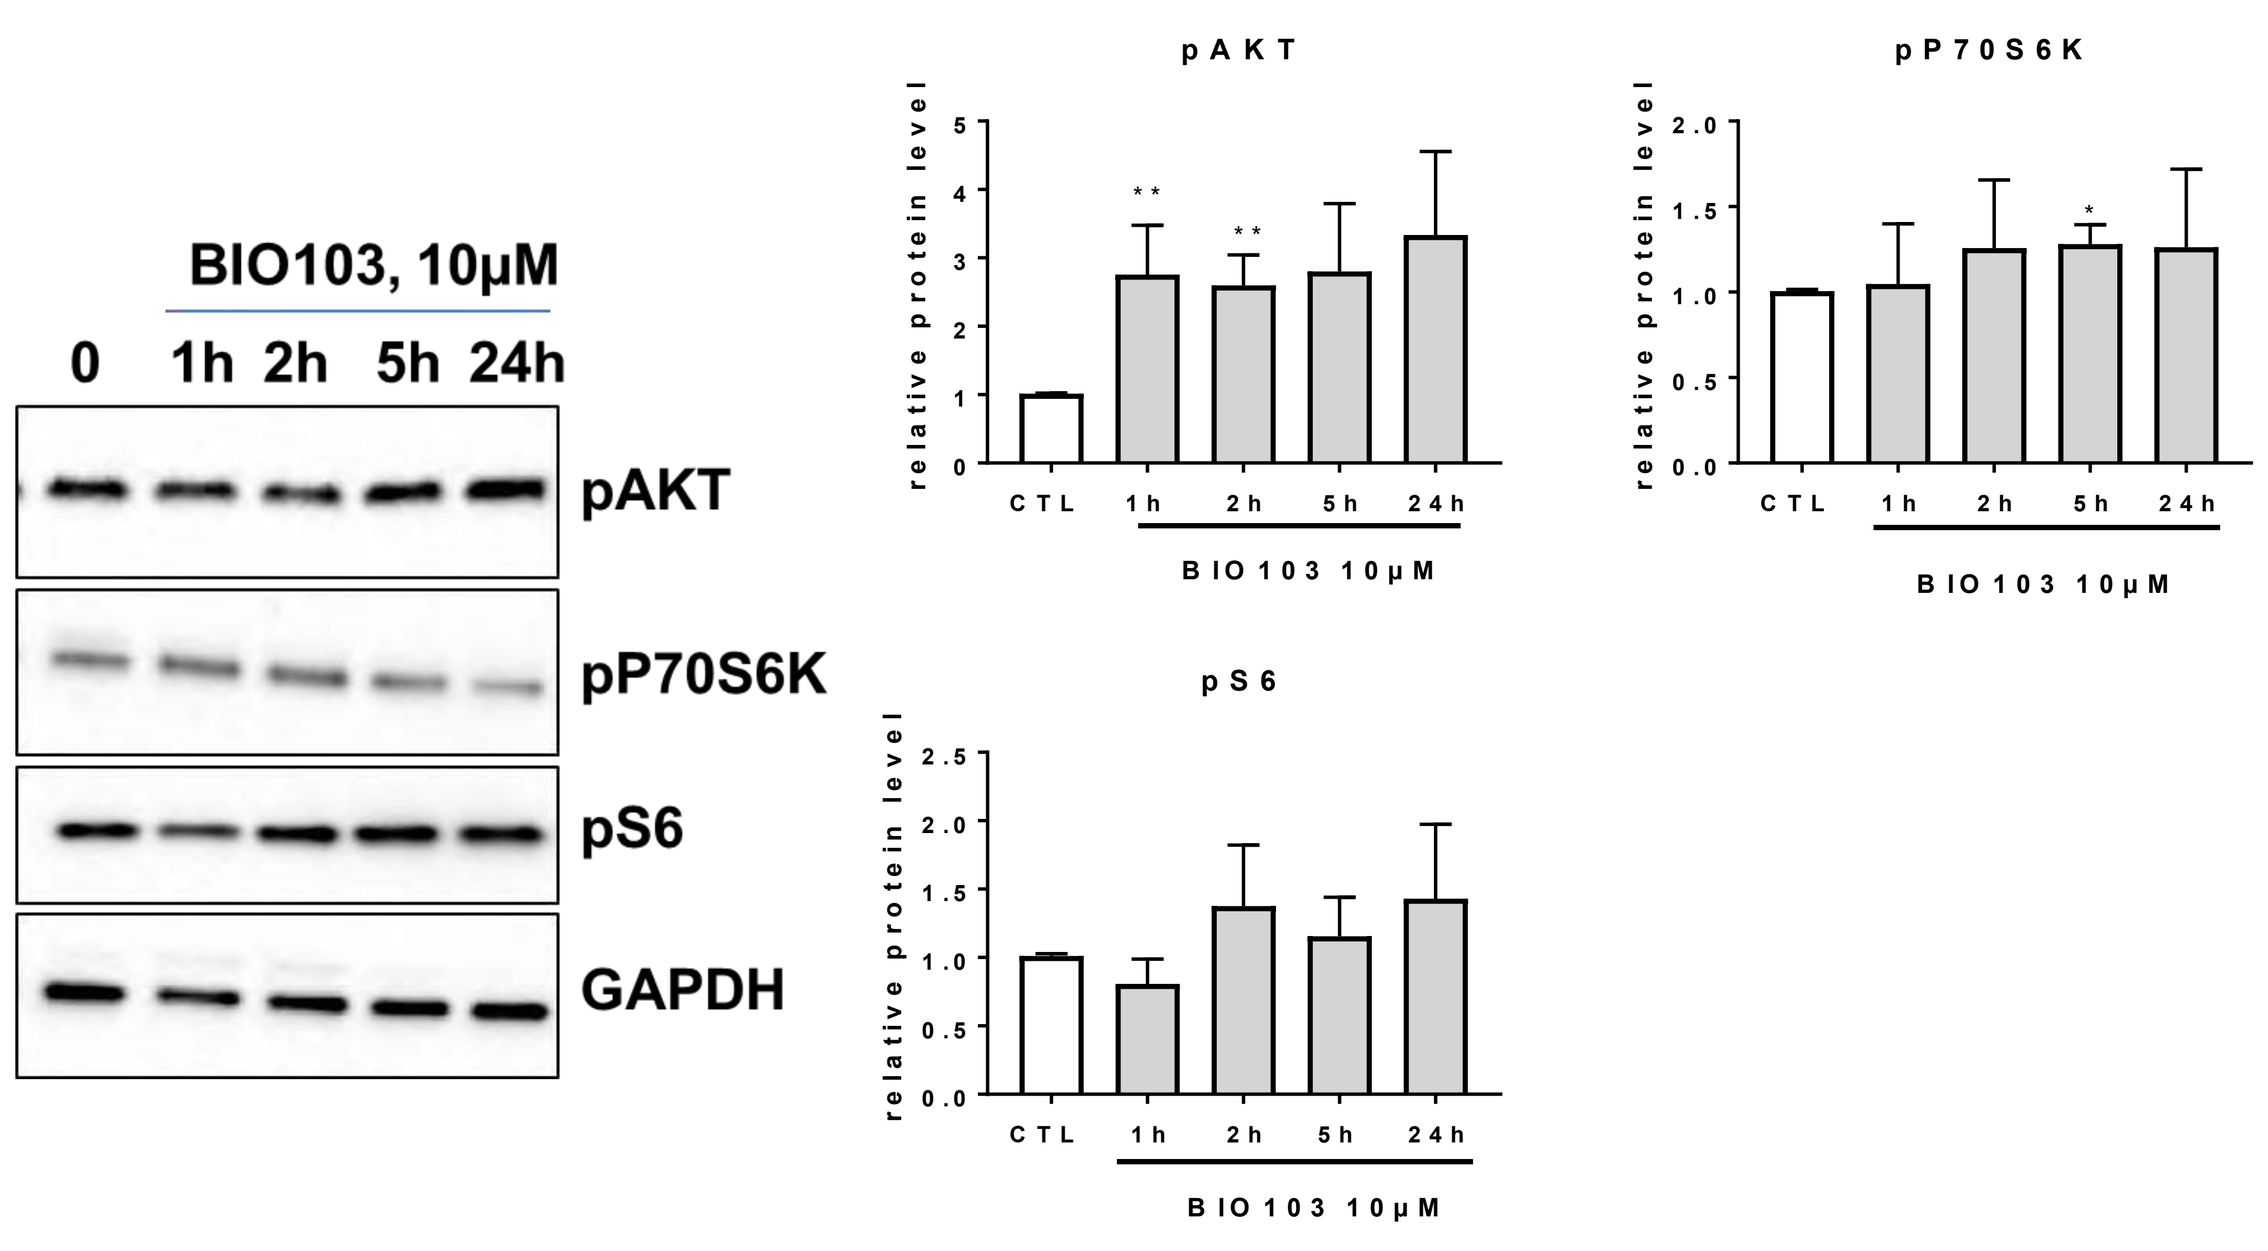

BIO103, 10µM
0
1h
2h
5h
24h
pAKT
pP70S6K
pS6
GAPDH
relative protein level
CTL
BIO103 10µM

- MAPK

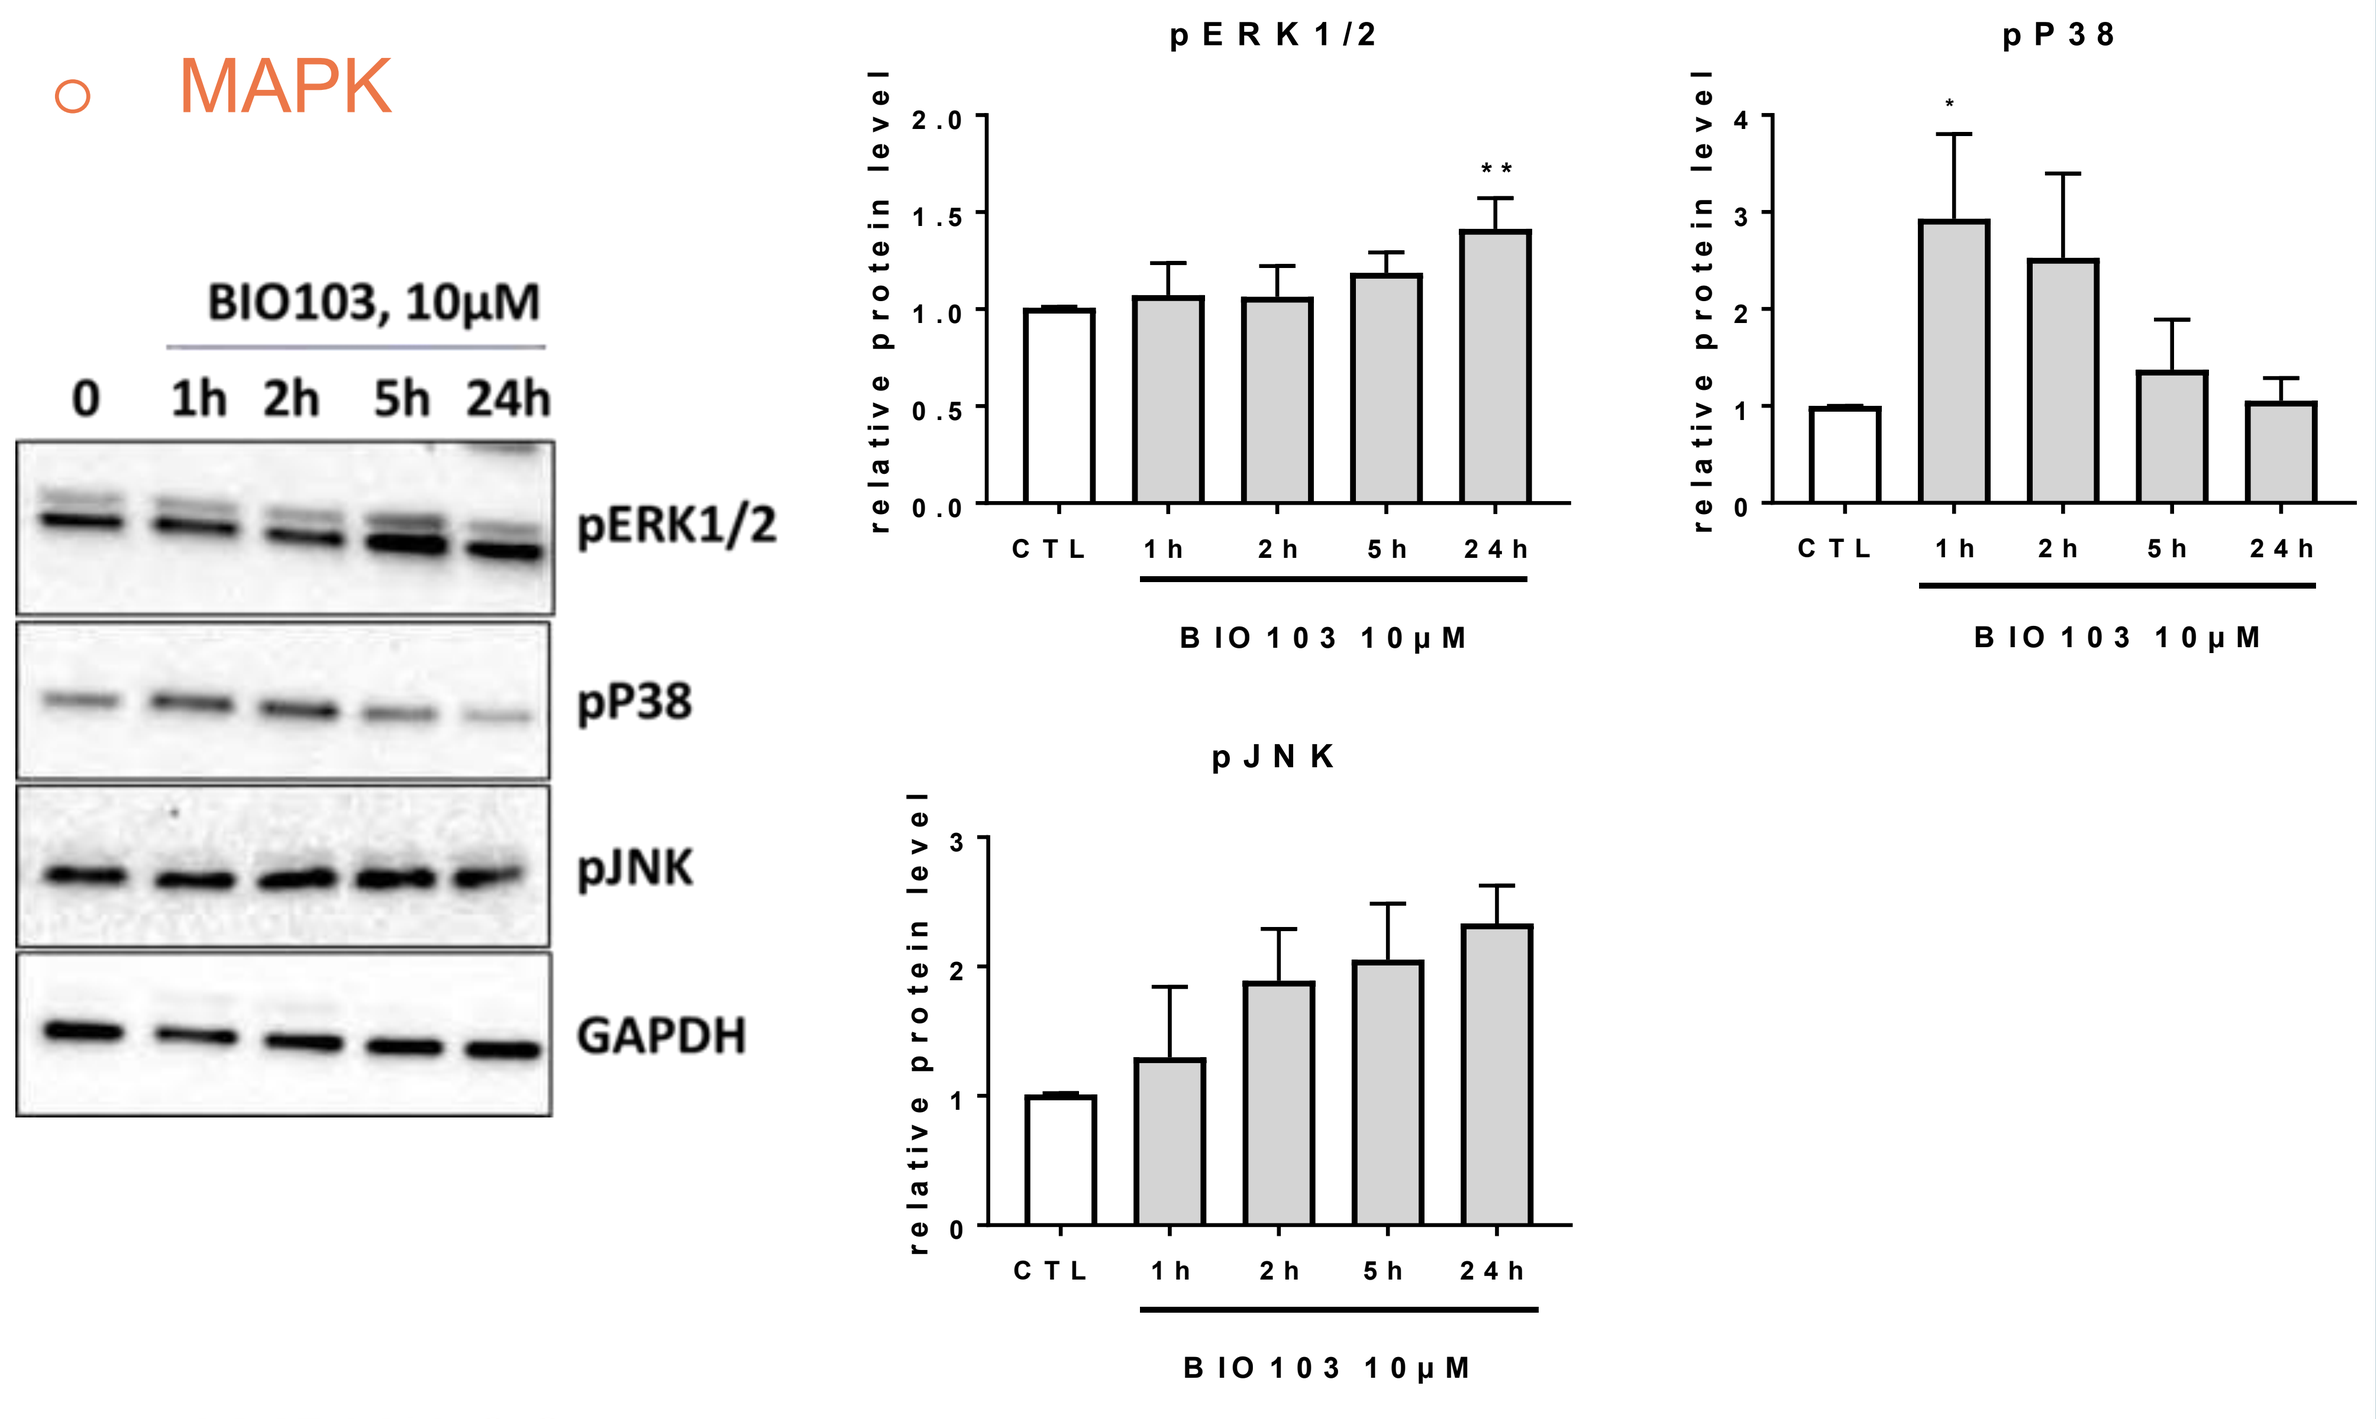

BIO103, 10µM
0
1h
2h
5h
24h
pERK1/2
pP38
pJNK
GAPDH
relative protein level
CTL
BIO103 10µM

- AMPK/ACC

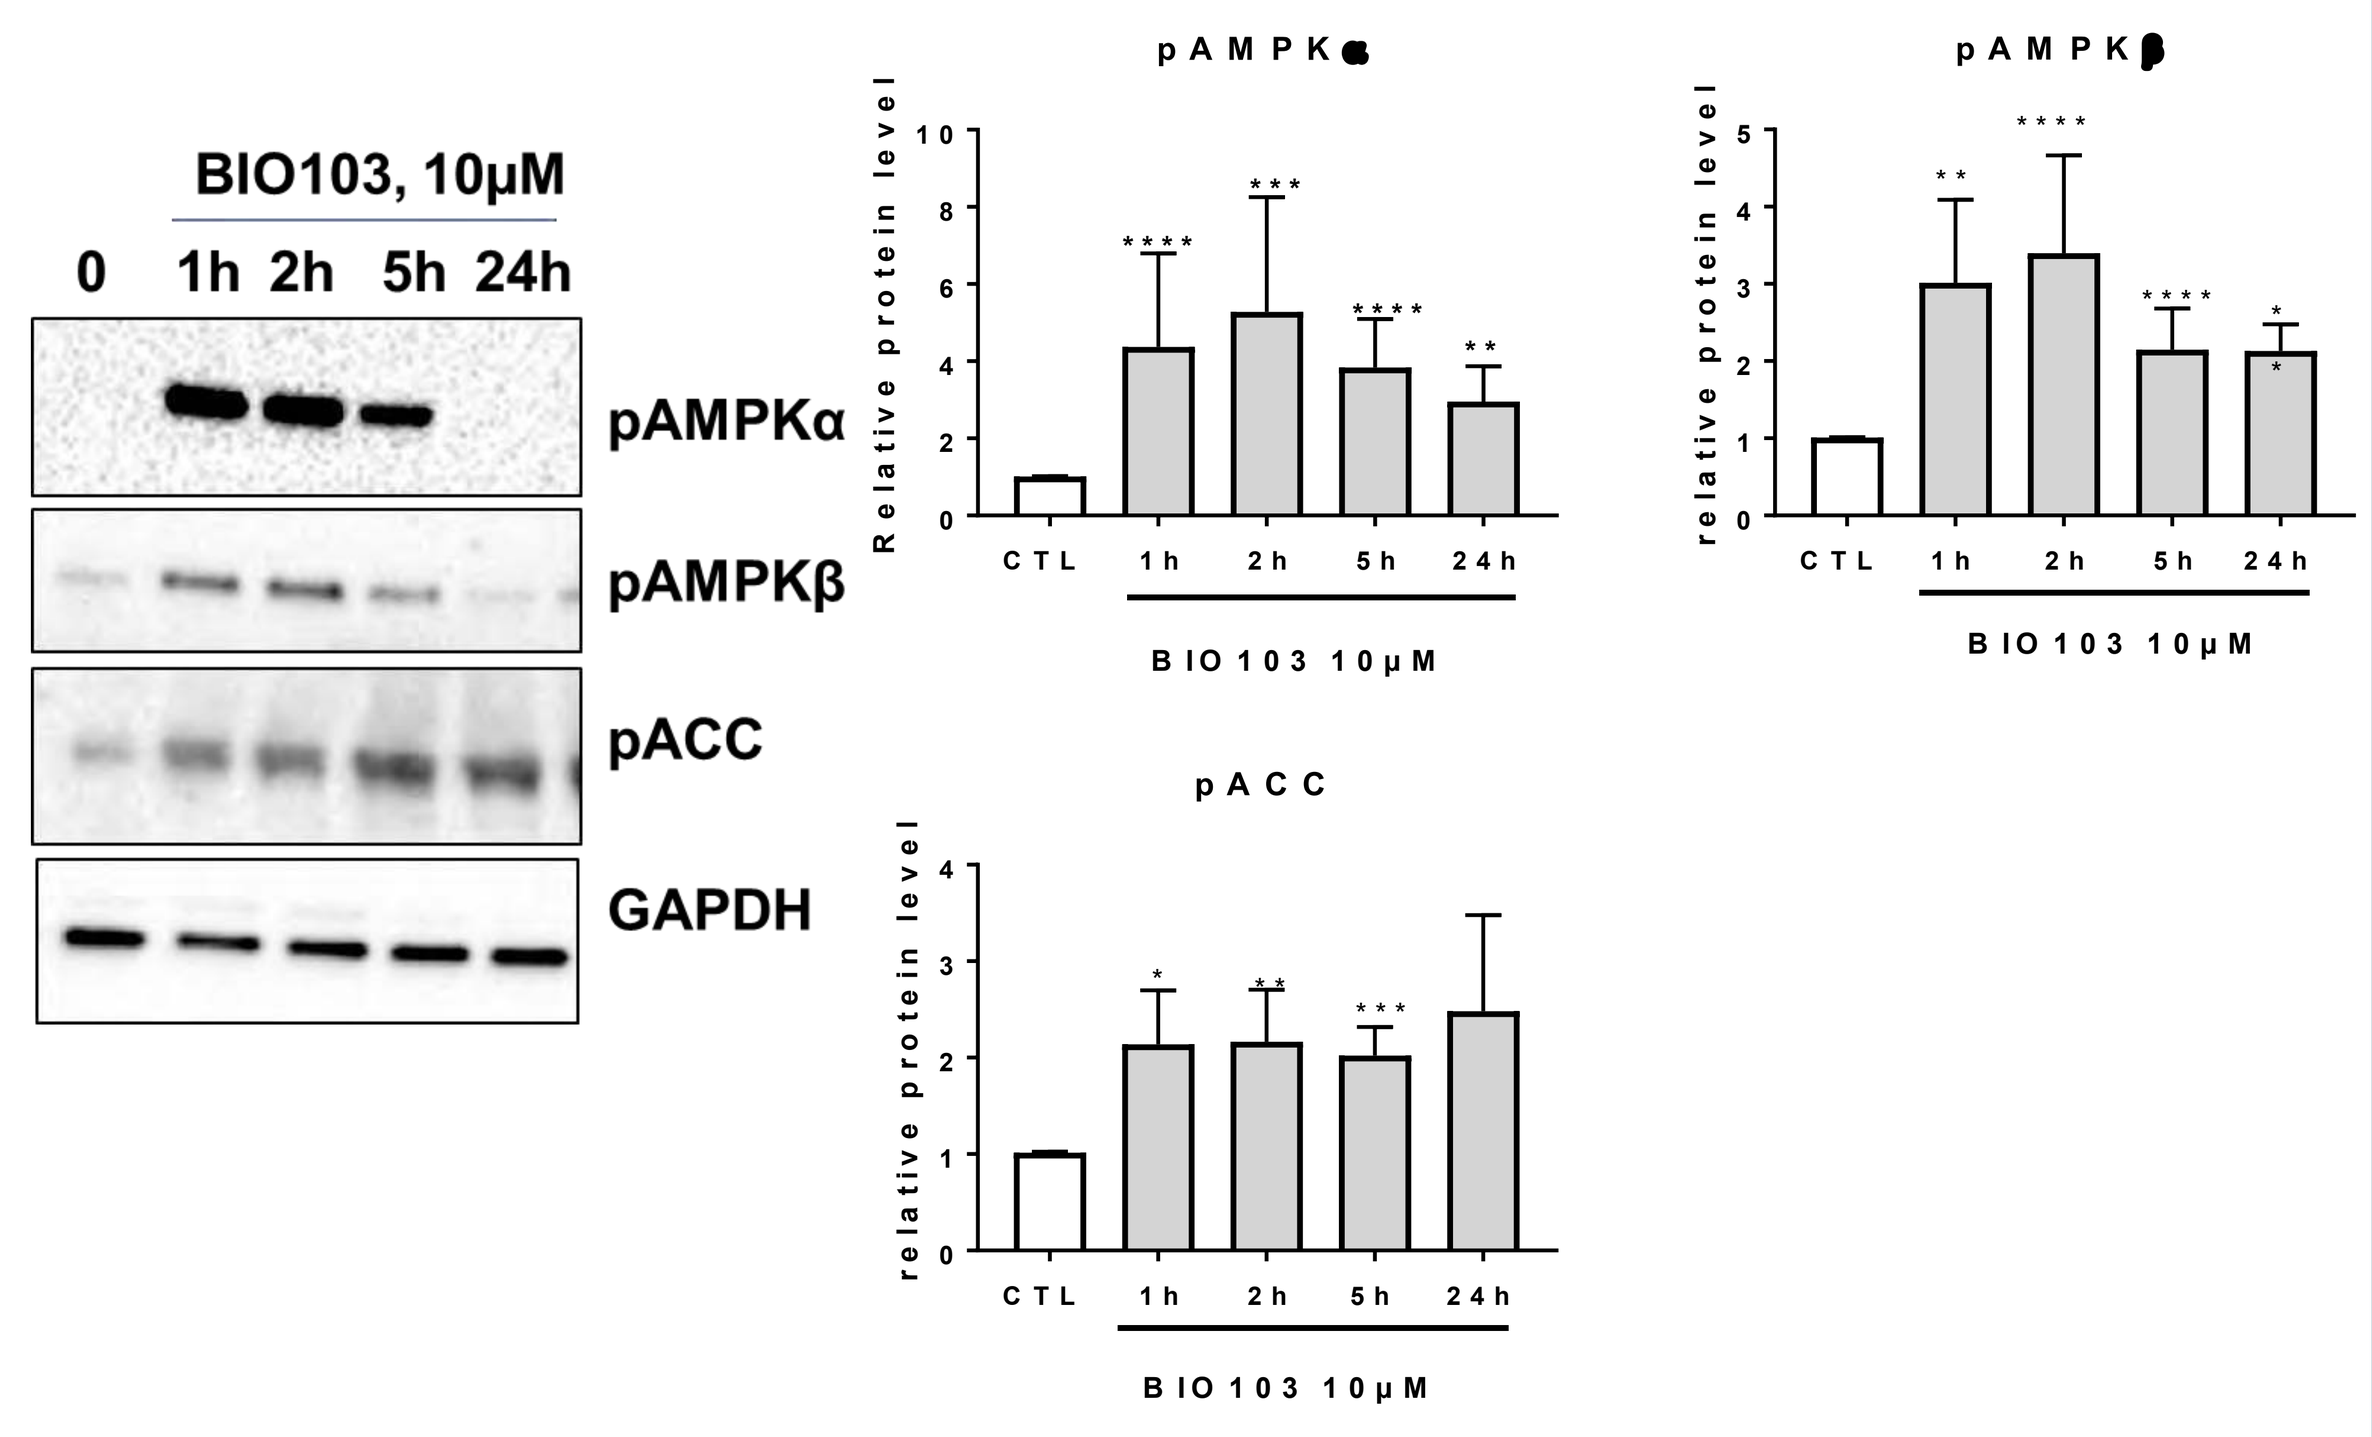

BIO103, 10µM
0
1h
2h
5h
24h
pAMPKα
pAMPKβ
pACC
GAPDH
Relative protein level
CTL
BIO103 10µM

→ BIO103 rapidly activates the major kinases of PI3K/AKT/mTOR, MAPK and AMPK signaling pathways in differentiated C2C12 cells

Effects of BIO103 on mitochondrial respiration and glycolysis of differentiated C2C12

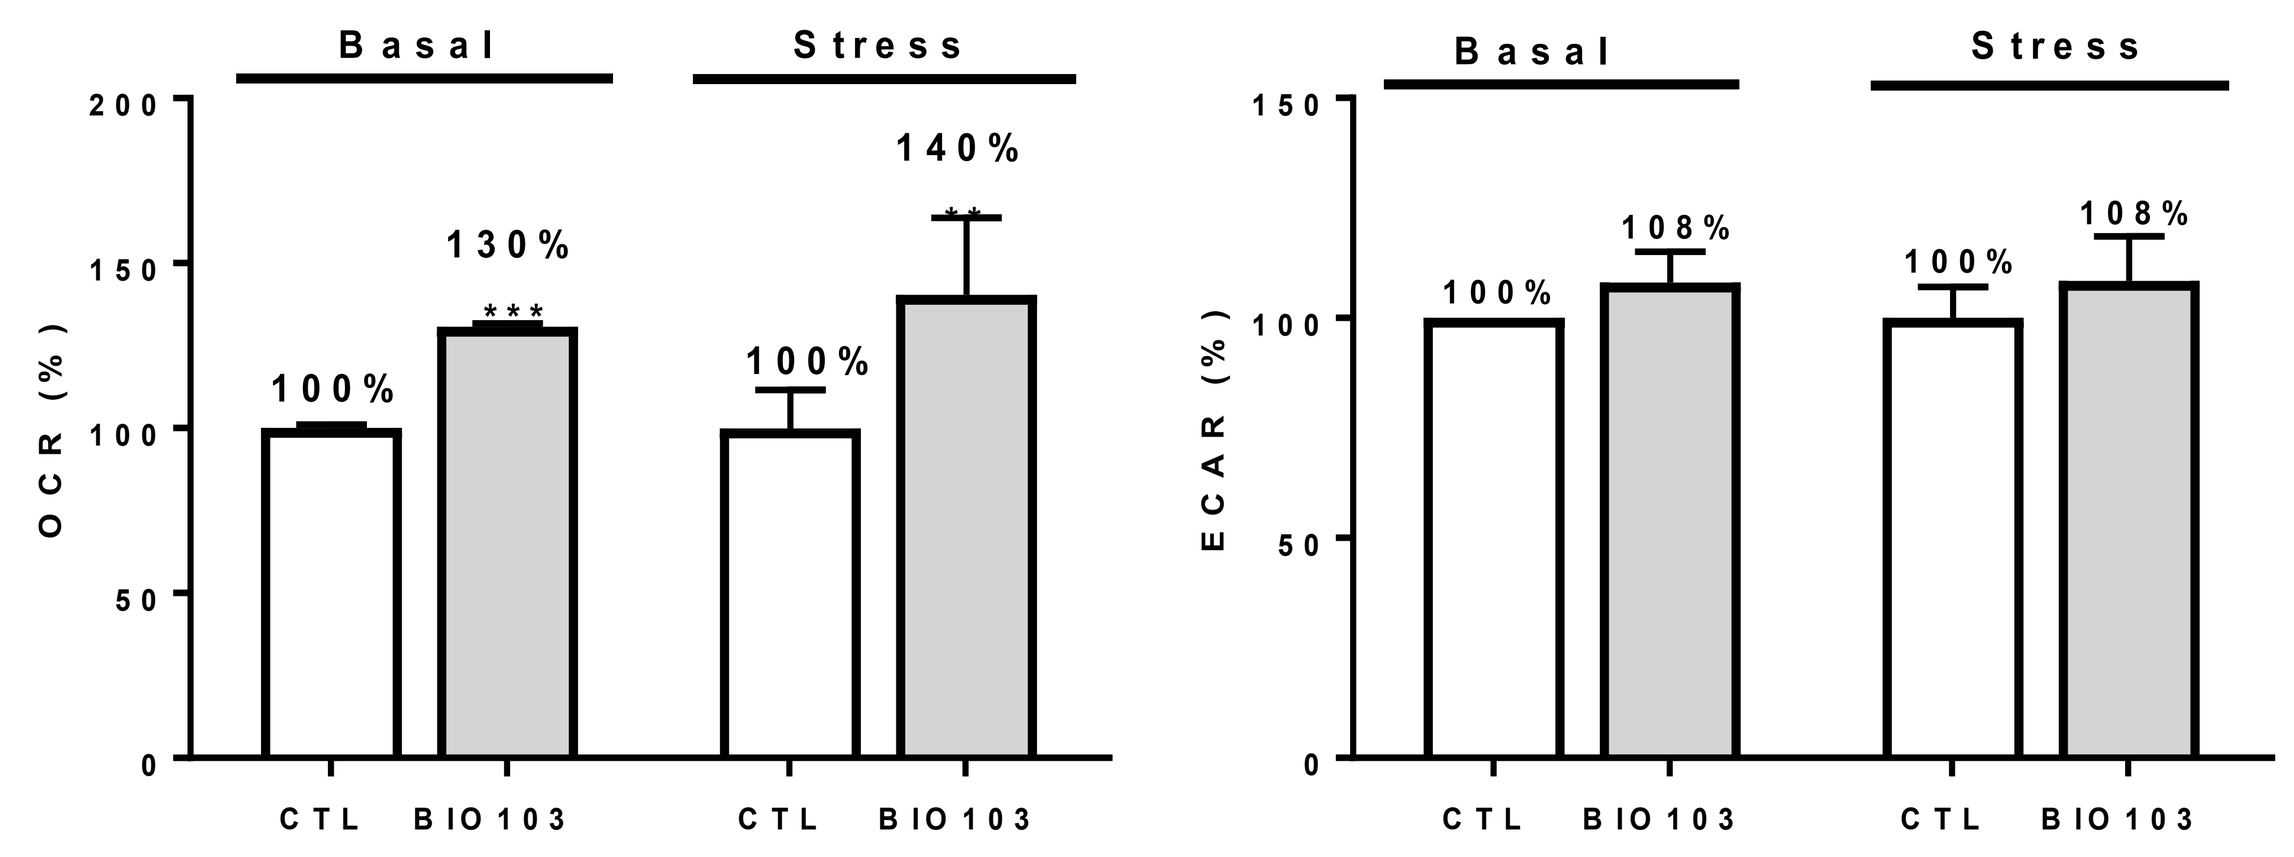

Basal
Stress
OCR (%)
ECAR (%)
100%
130%
140%
108%
CTL
BIO103

→ BIO103 increases both basal and maximal oxygen consumption rates (OCR) in muscle C2C12 cells after 72 h exposure to differentiated cells

- **In vivo**

Old (22 months) C57Bl6/J female mice were treated orally for 14 weeks with either vehicle or or BIO103 (50 mg/kg/day).

Running velocity

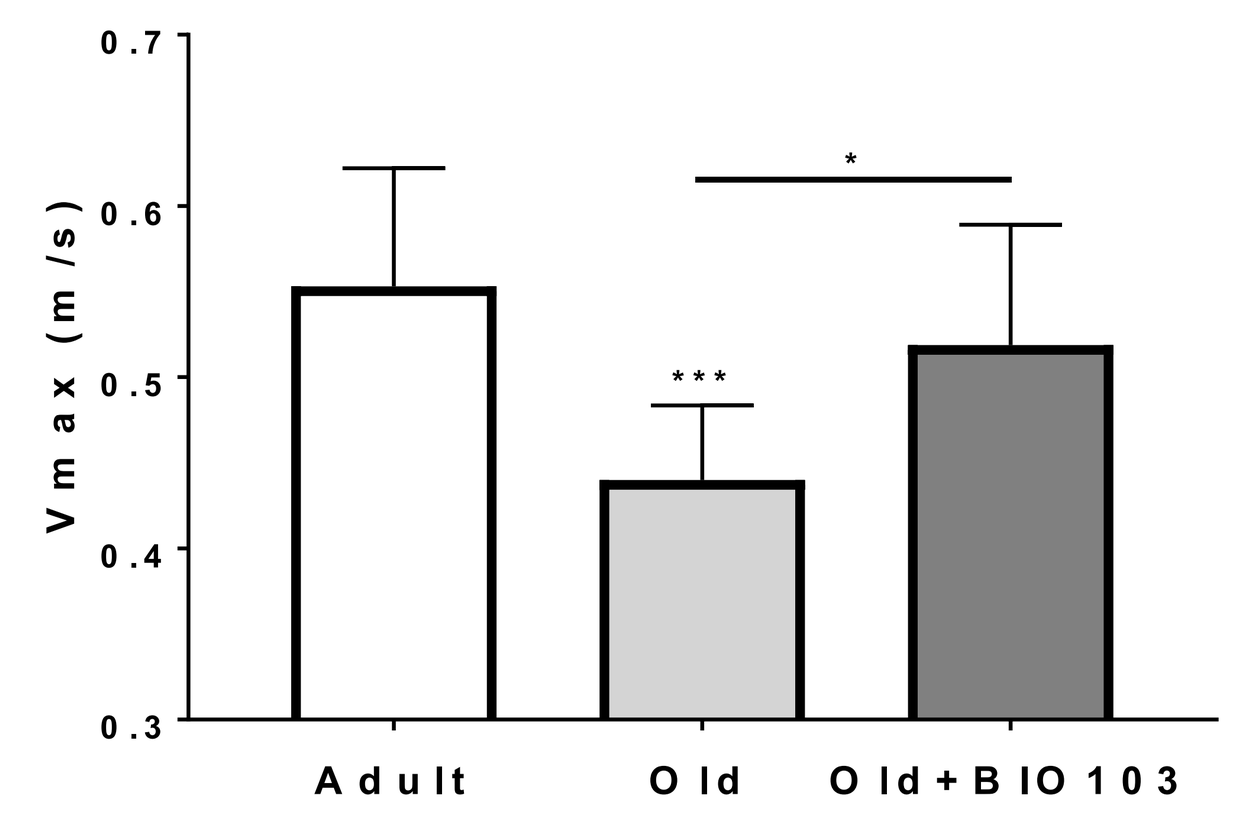

Vmax (m/s)
Adult
Old
Old+BIO103

→ Treatment of old animals by BIO103 compensate for the significant loss of running velocity as a consequence of aging.

IGF-1

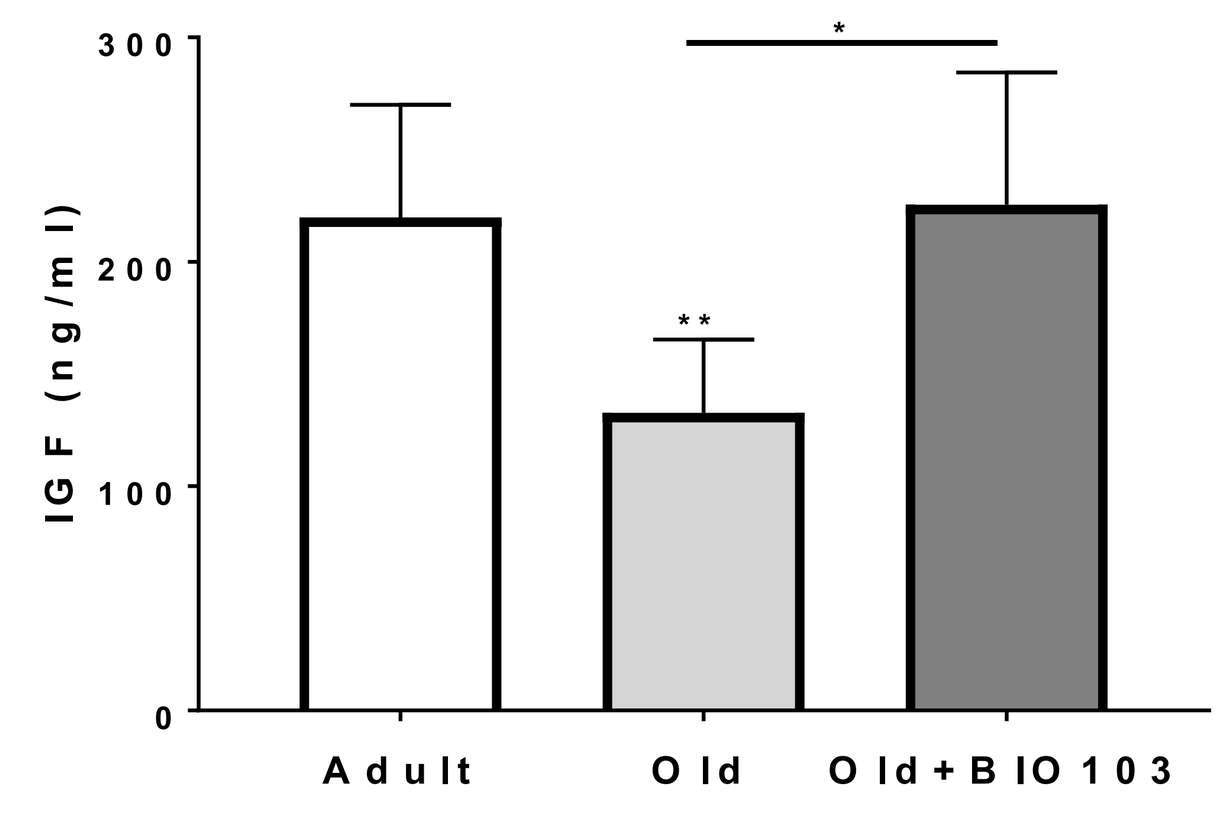

IGF (ng/ml)
Adult
Old
Old+BIO103

Myostatin

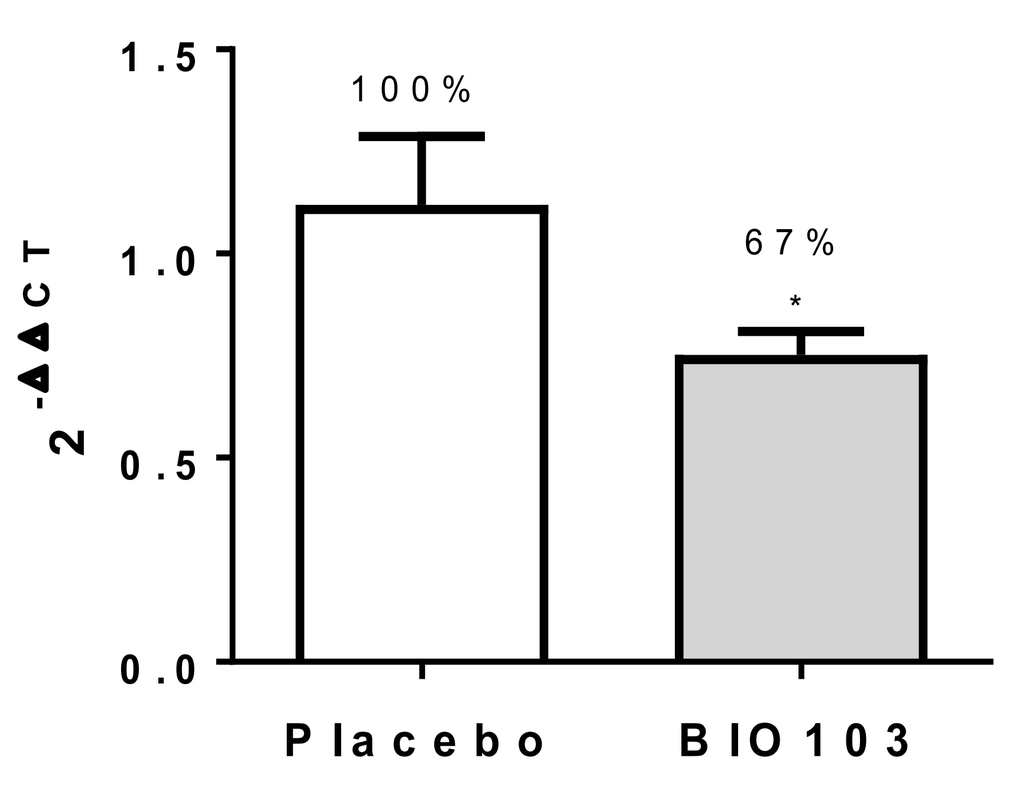

2-ΔΔCT
100%
67%
Placebo
BIO103

→ Chronic oral administration of BIO103 was responsible for a significant increase in animal IGF-1 plasma level and decreased myostatin expression in gastrocnemius muscles.

AMPK

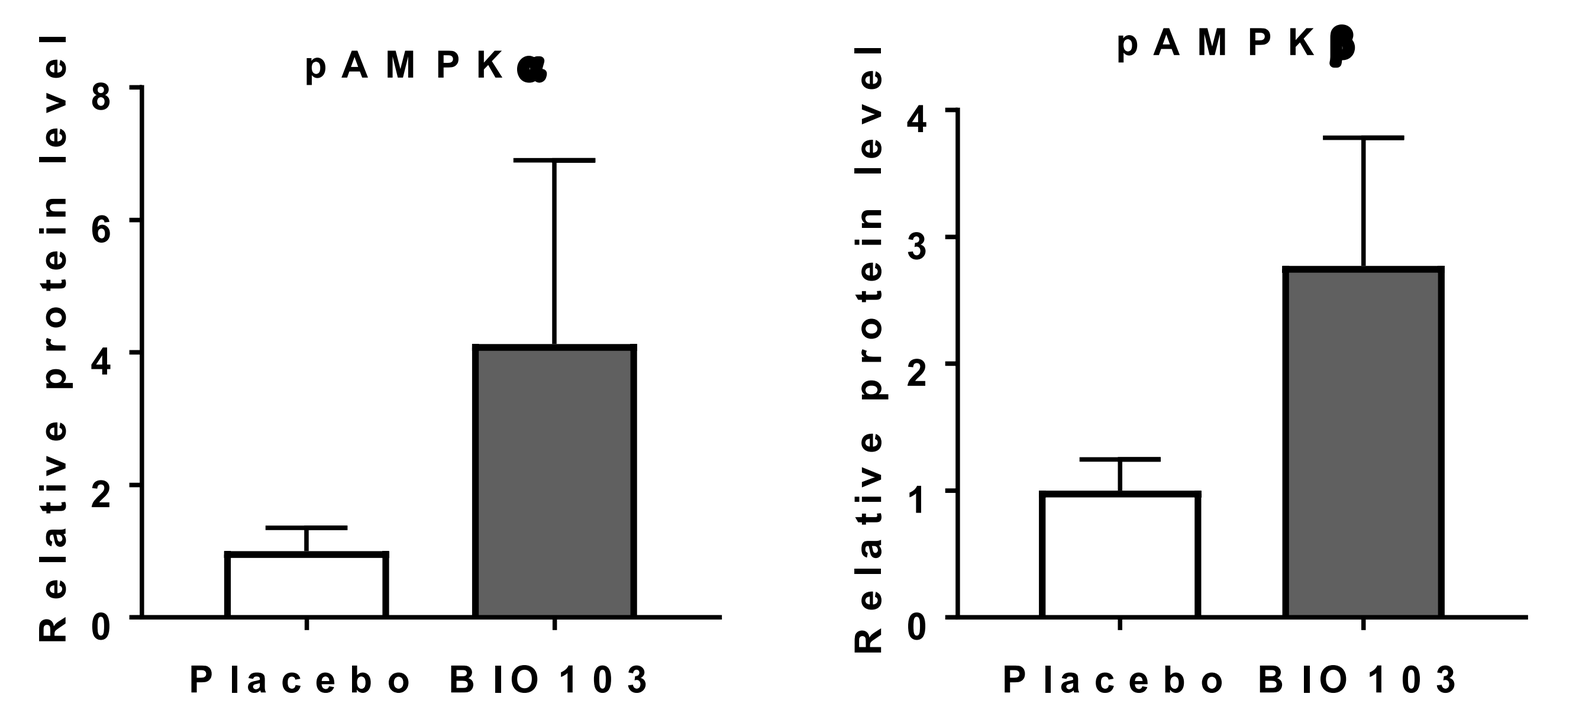

Relative protein level
Placebo
BIO103

→ BIO103 treatment tends to increase AMPK phosphorylation in gastrocnemius muscles of old mice

Conclusions

- **BIO103 displayed *in vitro* and *in vivo* anabolic effects on myofibers. These effects were accompanied with Myostatin inhibition.**
- **Anabolic properties of BIO103 result from an activation of AKT/mTOR, MAPK and AMPK pathways.**
- **BIO103 warrant further studies towards its development as a drug candidate for the treatment of muscle wasting disorders.**