BIO103, a second-generation compound for the treatment of sarcopenia: from anabolic properties to the reversion of aging-related functional loss

Pierre Dilda[1], Anne-Sophie Foucault[1], Sophie Raynal[1], Christel Carbonne[2], Jean-Denis Durand[2], Stanislas Veillet[1], Waly Dioh[1], René Lafont[3]

[1] Biophytis, BC9, 4 place Jussieu, 75005 Paris, France
[2] Metabrain Research, 4 Avenue du Président François Mitterrand, 91380 Chilly-Mazarin, France
[3] Sorbonne Universités, UPMC Univ Paris 06, CNRS - Institut de Biologie Paris Seine (BIOSIPE) 75005 Paris, France

Background

Muscular degeneration or sarcopenia is a natural process which accelerates with age. It is characterised by a loss of skeletal muscle mass and function, which is at the origin of a general deterioration in physical condition. Muscle atrophy results from a reduction of fibre number and diameter. Obesity aggravates sarcopenia and impairs functional capacities. The aim of this study was to characterize a new hemi-synthetic derivative of 20-hydroxyecdysone (BIO103) *in vitro* on myocytes and *in vivo* on various animal models designed to analyze the effects of high-fat diet, aging and muscle disuse.

Methods

Murine C2C12 cells were employed to evaluate the effects of BIO103 on protein synthesis (via ELISA measurements of pS6K levels) and on myostatin and atrogin gene expression (qRTPCR). The oral bioavailability of BIO103 was determined in rats (n=3) who received a single dose of the compound either oraly (50 mg/kg) or intraveously (5 mg/kg). Plasma concentrations were determined by HPLC-MS/MS at various time points and AUC were then calculated for each route of administration. In vivo, two animal models employing C57BL/6J mice under high-fat diet (4 442 kcal/ kg; proteins 19.8%, lipids 23% and carbohydrates 39.5%) were used. The first one involved 12 week-old males (n=8) treated orally for 6 weeks with vehicle or BIO103 at 5 or 50 mg/kg/day. At completion of experimentation, the *soleus* of each animals was weighed and then tested for protein content and myostatin, myogenin and MyoD gene expression (qRTPCR). The second animal model compared old (22 months) versus adult (12 months) female mice treated orally for 14 weeks with either vehicle or 50 mg/kg/day of BIO103 (n=7-10). One week before the completion of the study, the animals were tested for functional activity *in toto* (maximal running velocity on treadmill) and their right hind limb was immobilized. At the end of the experimentation, *in situ tibialis anterior* contractility parameters (maximal force and fatigue resistance) were recorded on both immobilized and active limbs. At euthanasia, plasma was collected and various muscles were weighed. IGF-1 plasma levels were determined by ELISA assay.

Results

in vitro evaluations

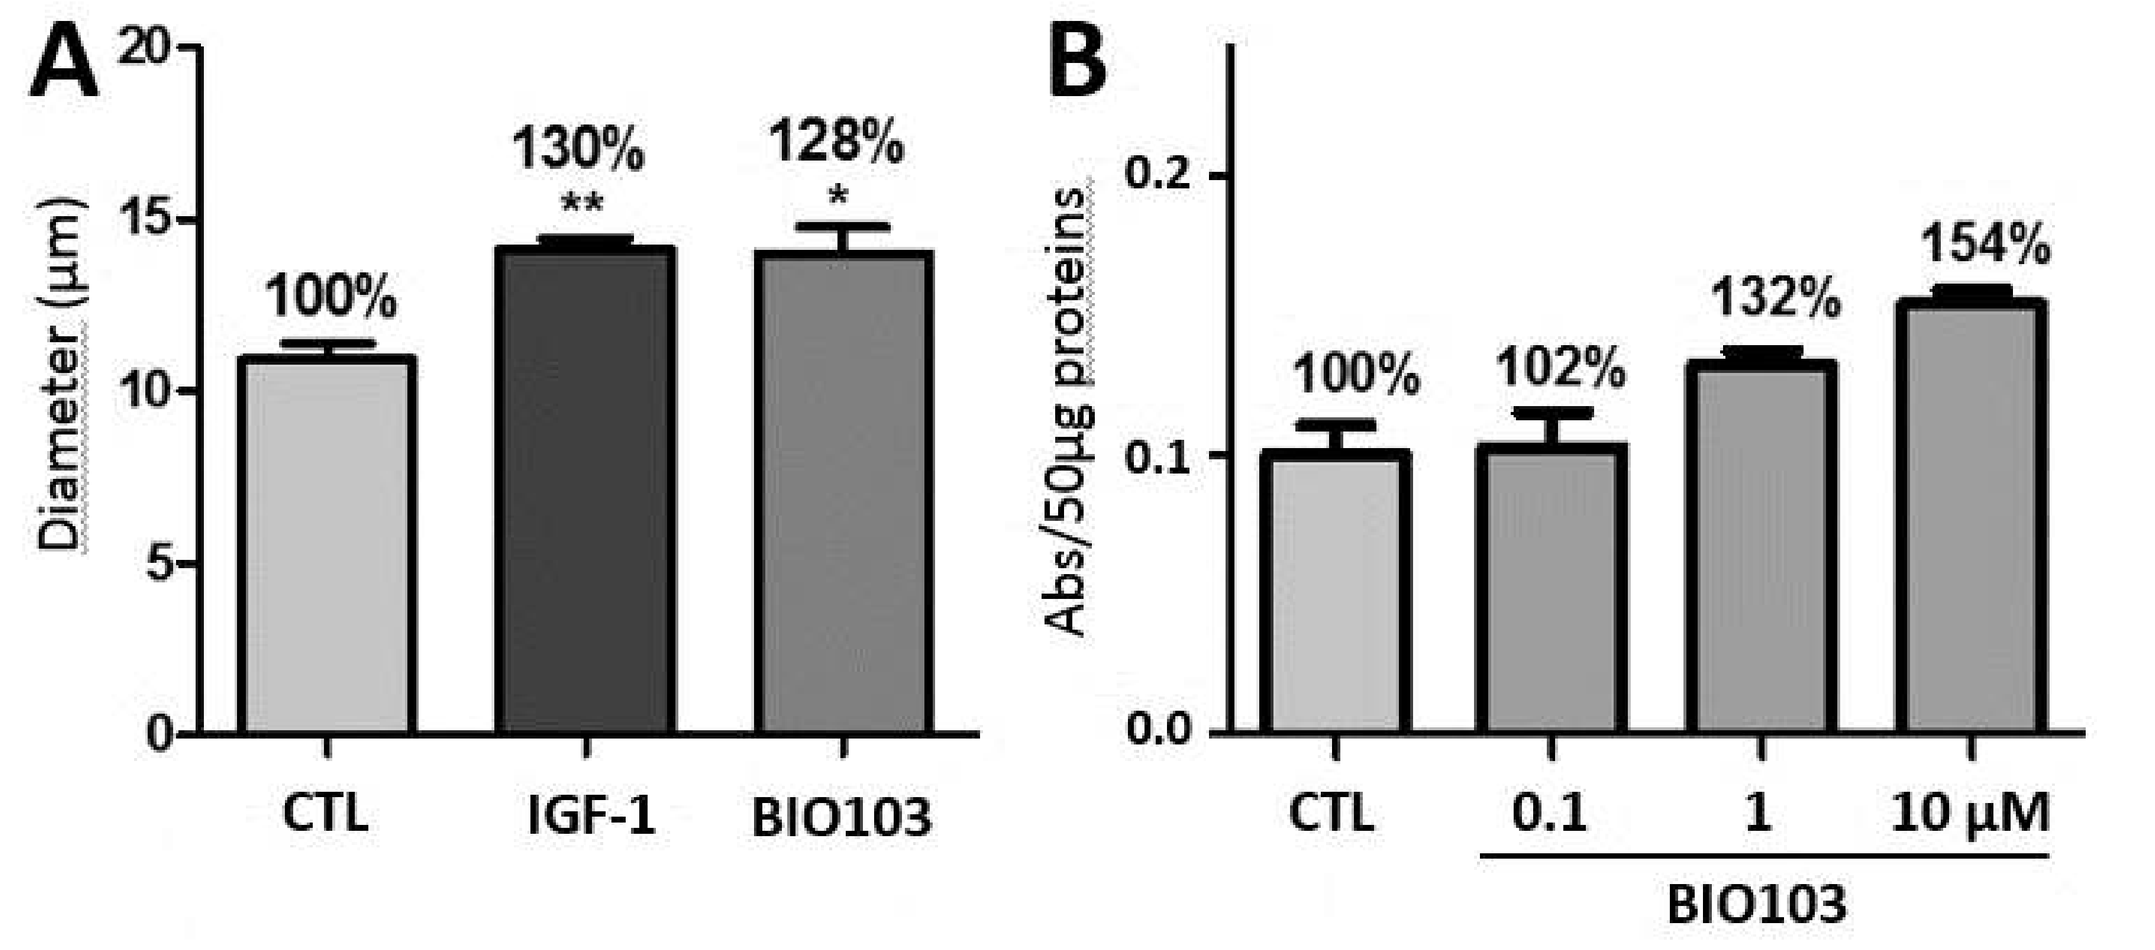
A
Diameter (µm)
100%
130%
**
128%
*
CTL
IGF-1
BIO103
B
Abs/50µg proteins
100%
102%
132%
154%
CTL
0.1
1
10 µM
BIO103

Figure 1: Effects of BIO103 on C2C12 differentiated cells. A/ Effect of BIO103 (1 µM) on C2C12 myotube diameter. B/ pS6K ELISA detection in cells exposed to 0.1 to 10µM BIO103. Statistical analysis was performed using an Anova followed by a Dunnett t test. *p<0.05, **p<0.01 vs control.

→ BIO103 is responsible for a significant increase in myotube diameter consistent with an activation of the mTOR signalling pathway.

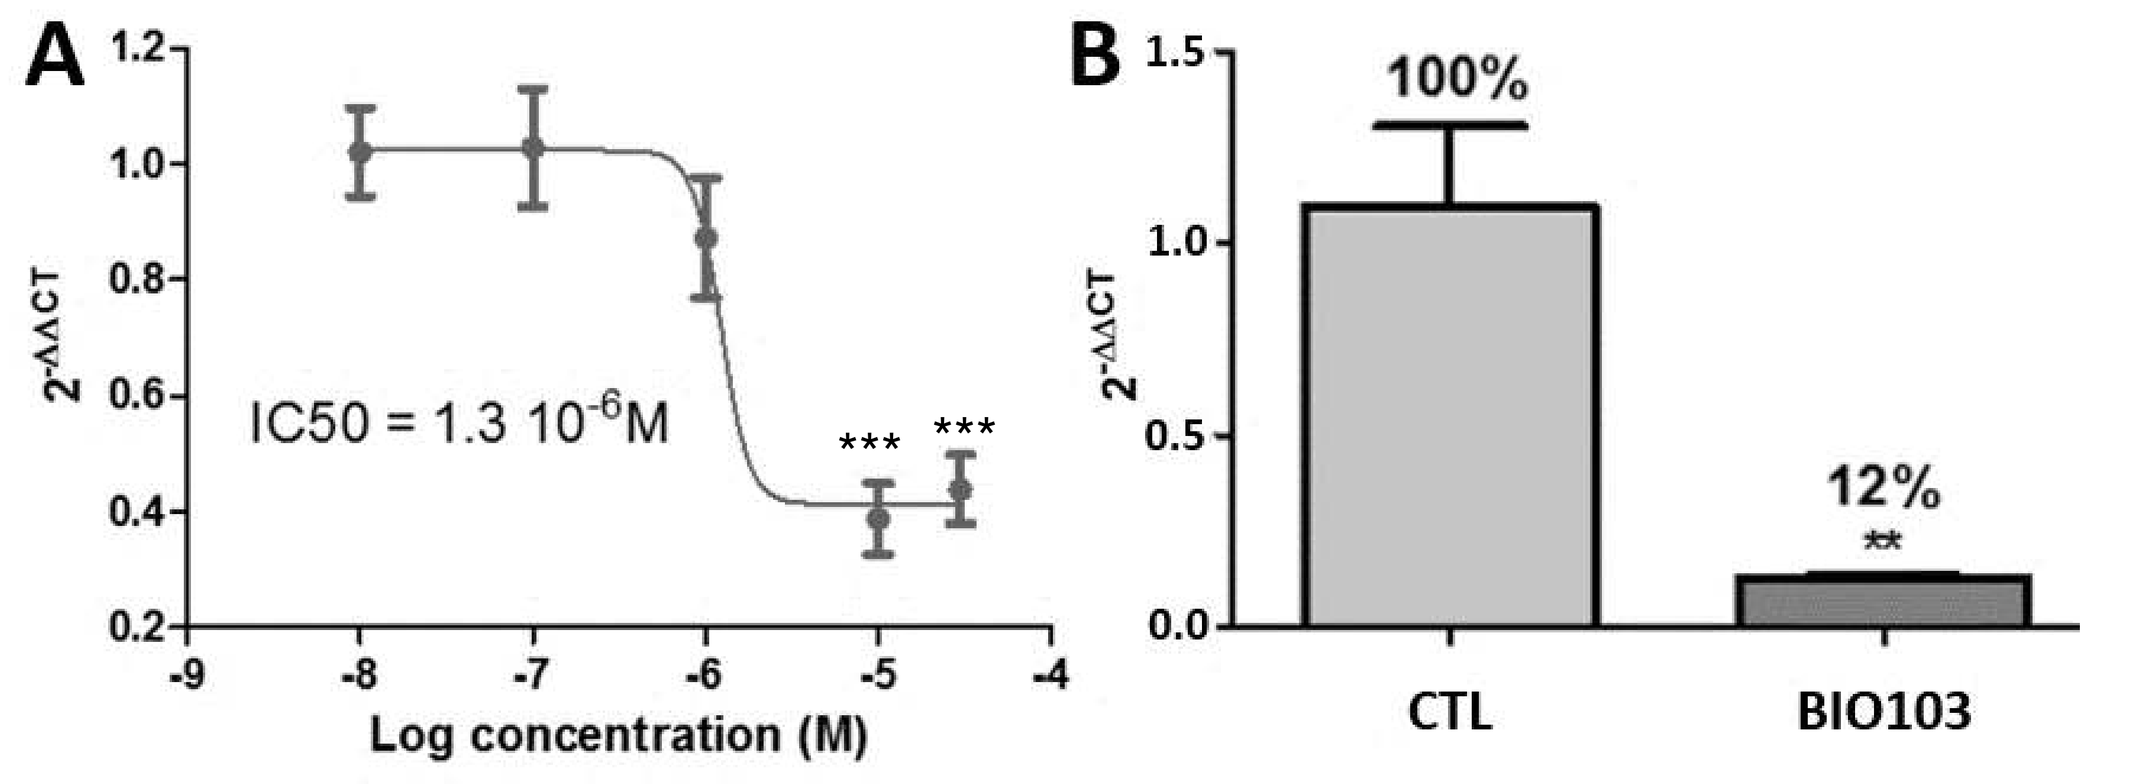
A
2^-ΔΔCT
IC50 = 1.3 10^-6M

Log concentration (M)
B
2^-ΔΔCT
100%
12%
**
CTL
BIO103

Figure 2: Effects of BIO103 on myostatin (A) and atrogin (B) gene expression. Differentiated C2C12 cells were treated with BIO103 for 6h. RNAs were extracted, purified and converted into cDNA to allow myostatin gene expression analysis by semi-quantitative PCR. The house-keeping gene used was beta-actin. Statistical analysis was performed using an Anova followed by a Dunnett t test. **p<0.01, ***p<0.001 vs control.

in vivo evaluations

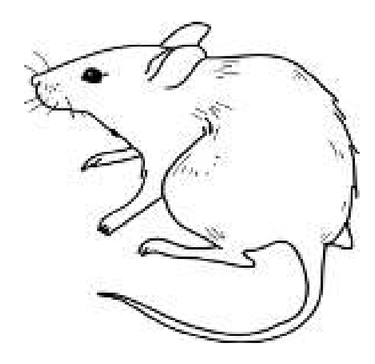

YOUNG ANIMALS

Ten-week C57Bl6/J male mice were treated orally for 6 weeks with either vehicle or BIO103 at 5 or 50 mg/kg/day under high-fat diet.

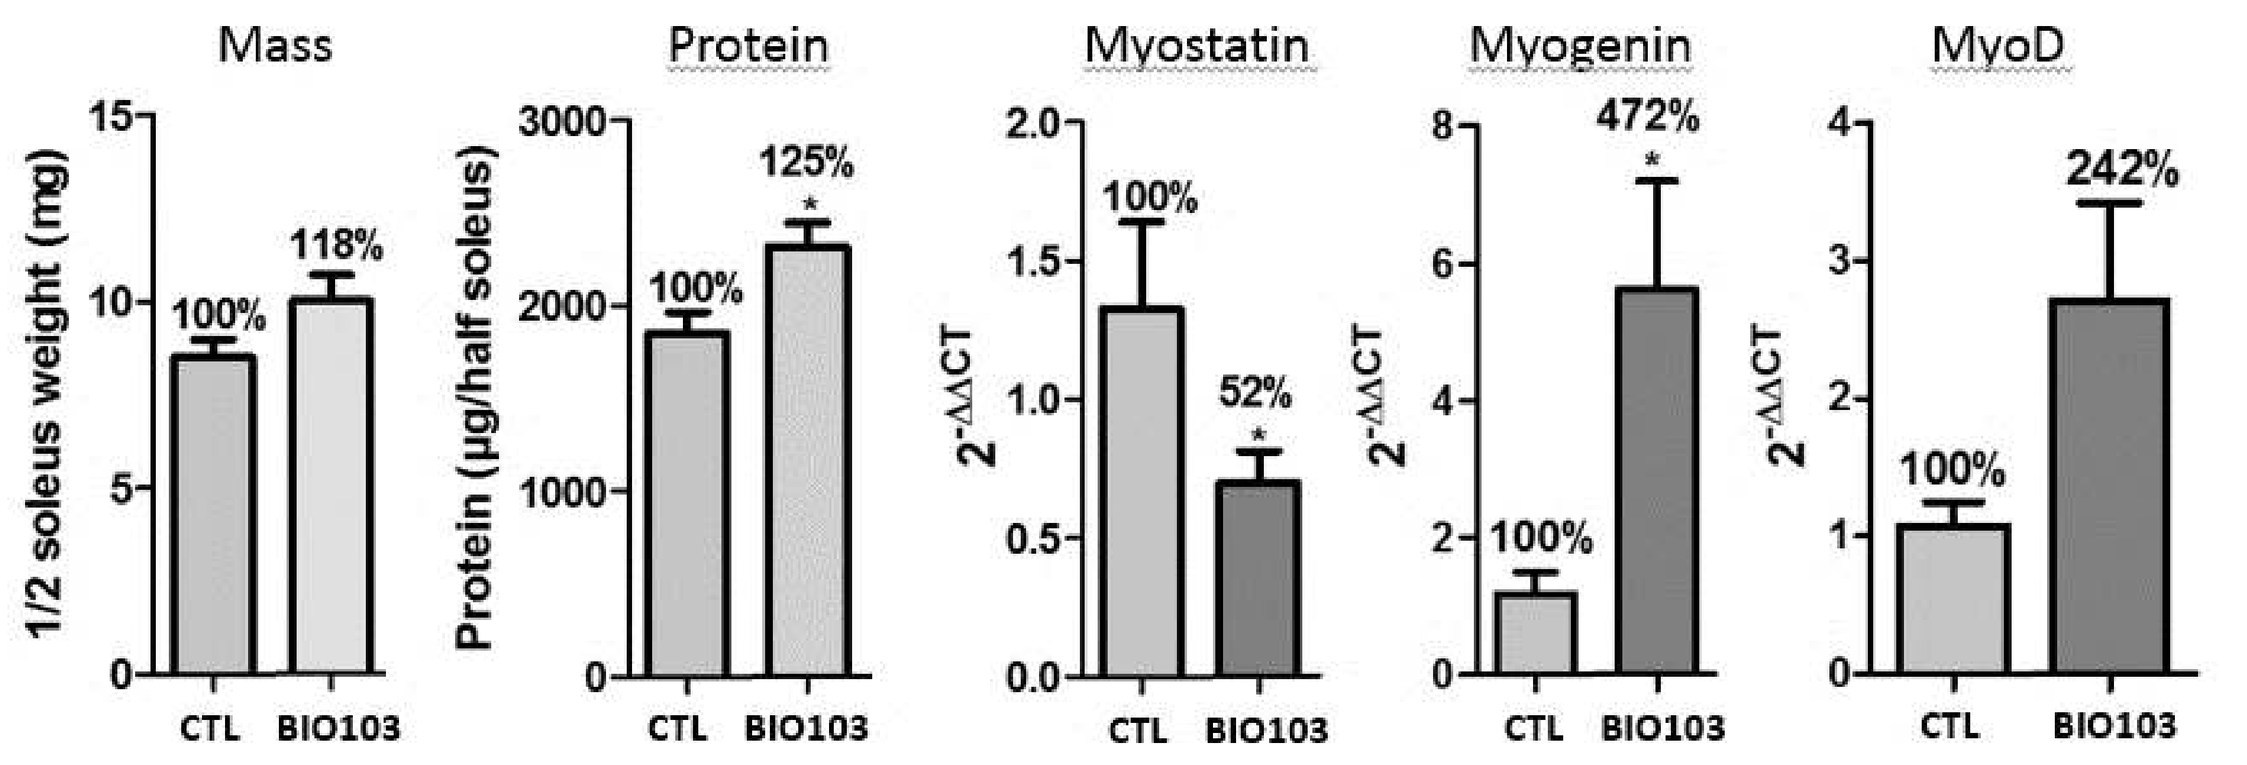
Mass
1/2 soleus weight (mg)
100%
118%
Protein
Protein (µg/half soleus)
100%
125%
*
Myostatin
2^-ΔΔCT
100%
52%
*
Myogenin
2^-ΔΔCT
100%
472%
*
MyoD
2^-ΔΔCT
100%
242%
CTL
BIO103

Figure 3: Effects of BIO103 on soleus muscle in young mice. Soleus weight and total protein content were determined in mice exposed with BIO103 at 5mg/kg per day. Myostatin and myogenesis transcription factor (myogenin and MyoD) gene expressions were determined in soleus muscle isolated from mice exposed to 50 mg/kg per day of BIO103.

→ BIO103 is responsible for a significant increase in muscle mass and protein content which is consistent with improved myogenesis transcription factor pattern.

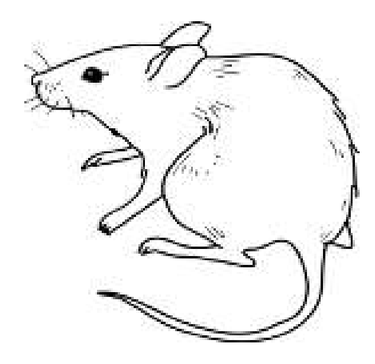

ADULT vs OLD ANIMALS

Adult (12 months) and old (22 months) C57Bl6/J female mice. were treated orally for 14 weeks with either vehicle or BIO103 (50 mg/kg/day). The adult mice received only the vehicle. All animals were subjected to a high fat diet for the duration of the experiment. One week before the completion of the experiment the right rear leg of the animals was immobilized.

Muscle weight

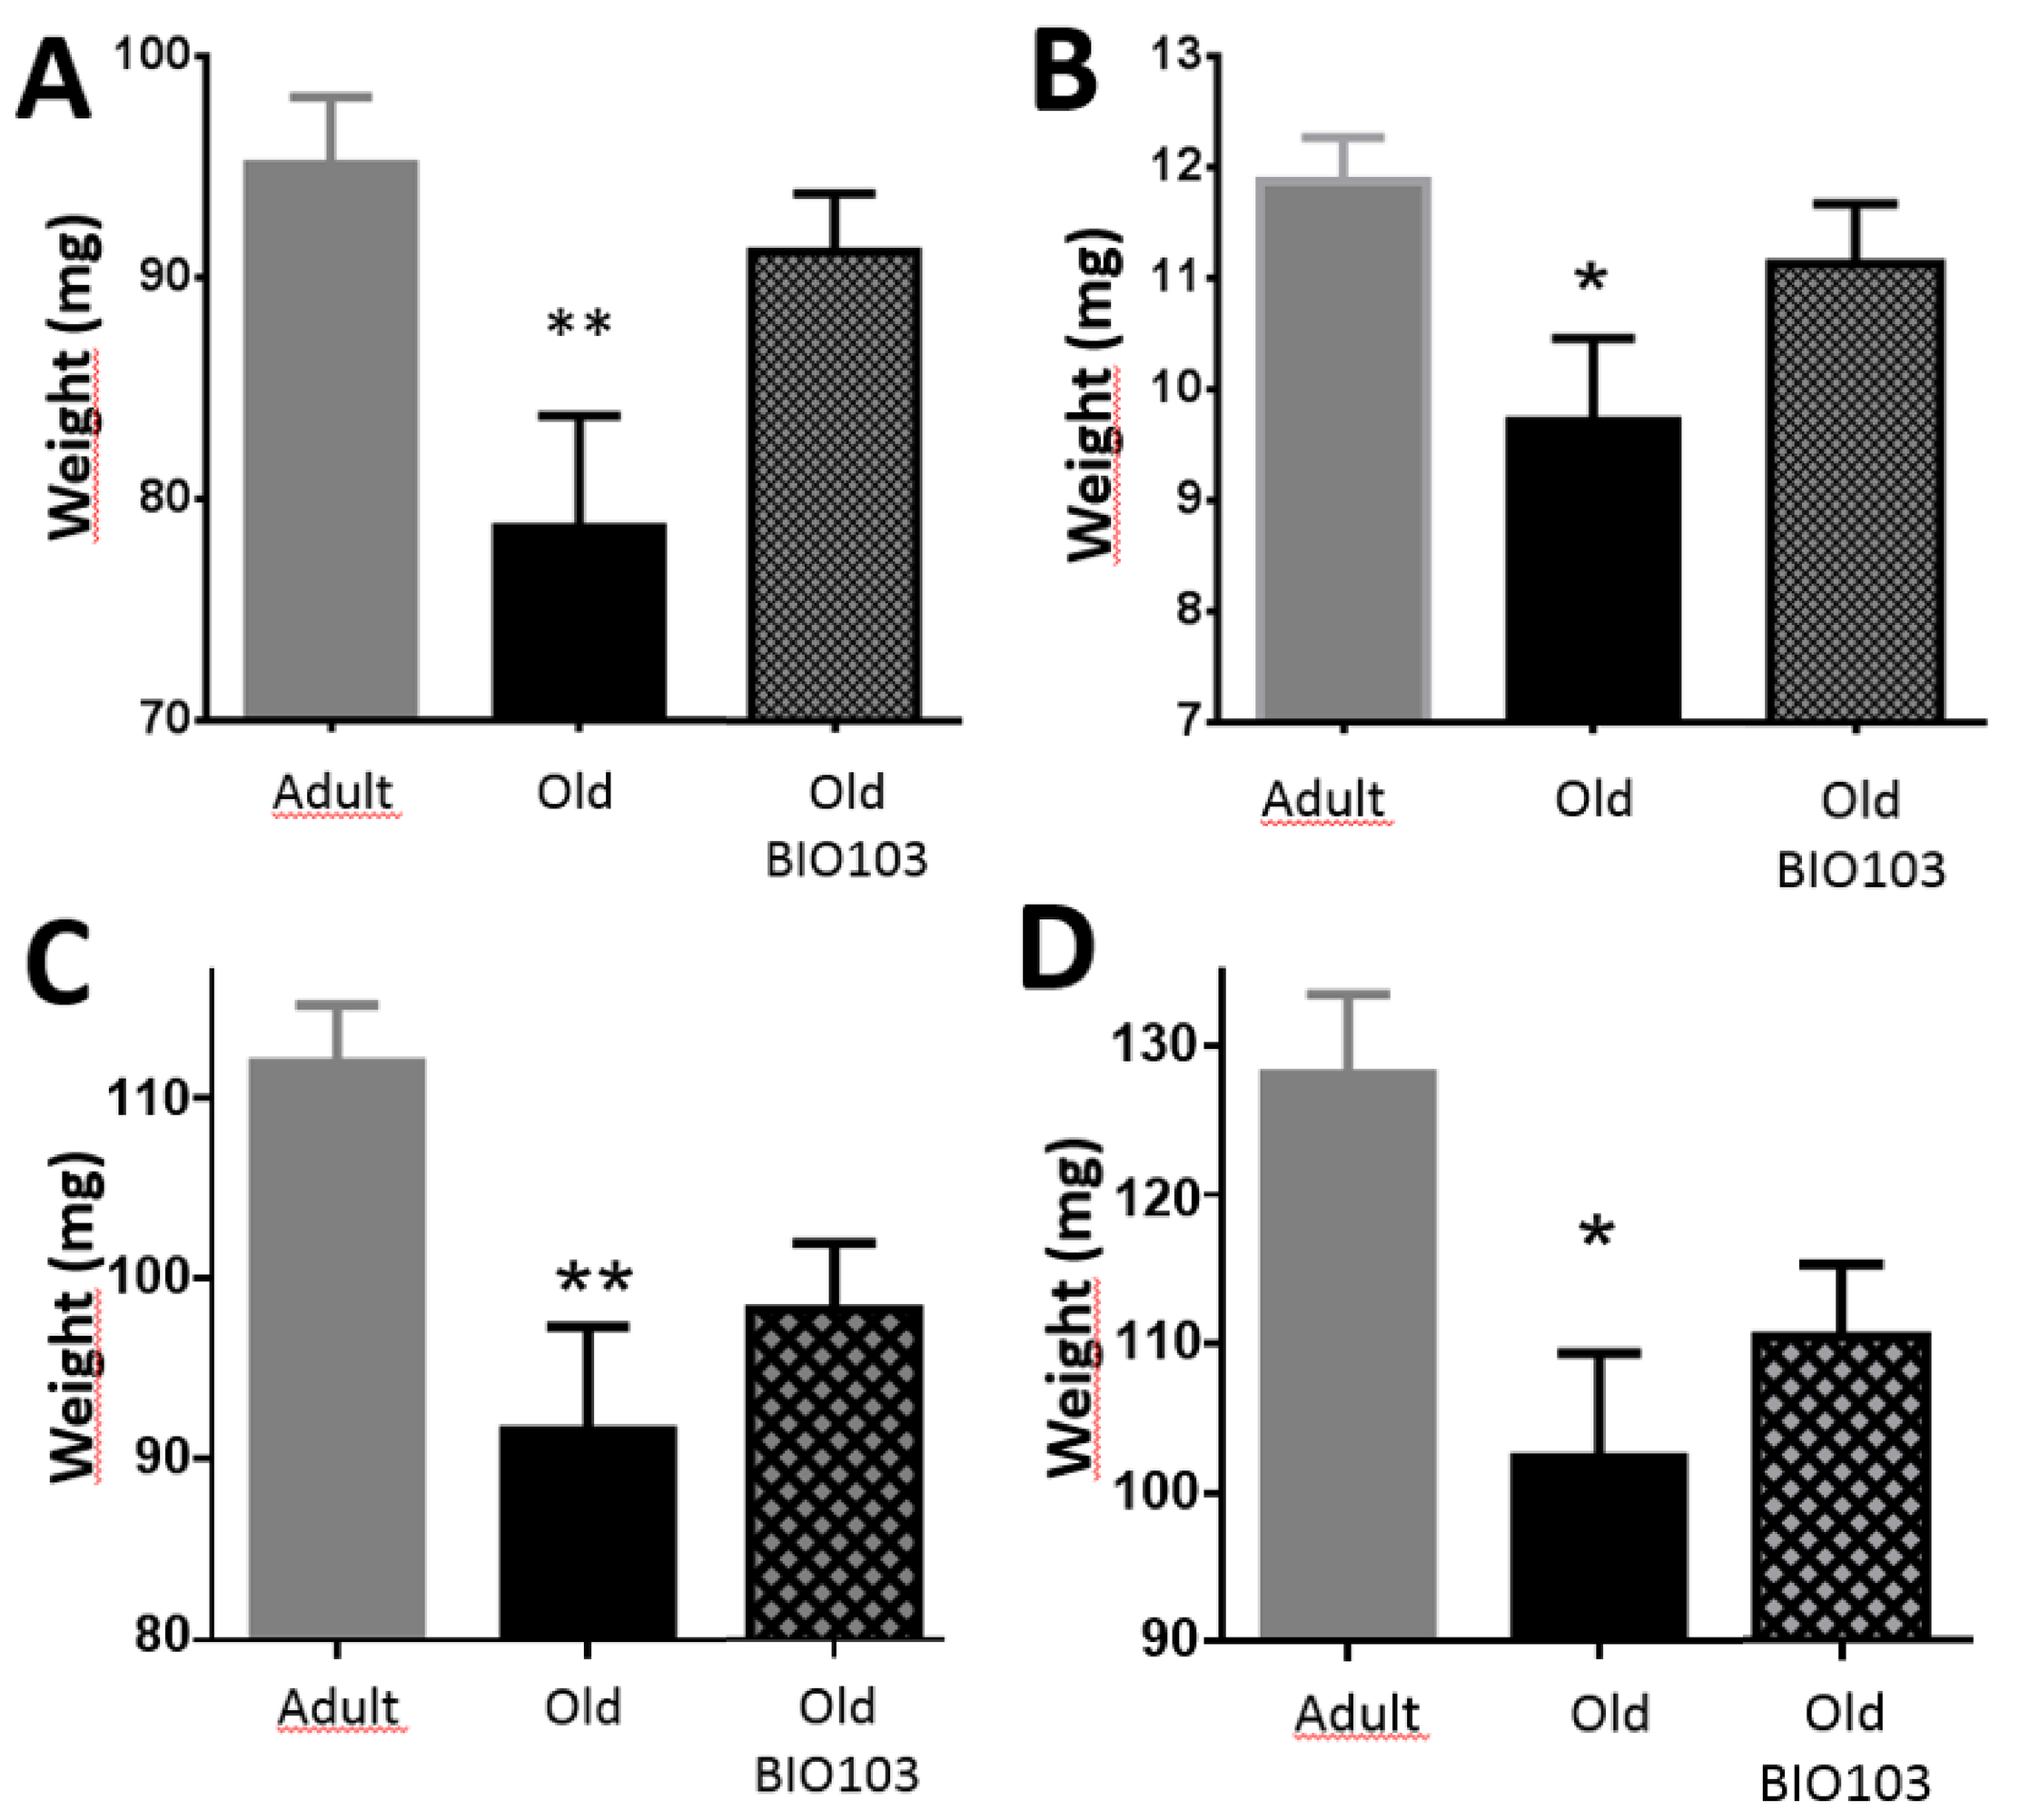
A
Weight (mg)
**
B
Weight (mg)
*
C
Weight (mg)
**
D
Weight (mg)
*
Adult
Old
Old BIO103

Figure 4: Effects of BIO103 on muscle weight in adult and old mice. Adult (12 months; n=9) and old (22 months) C57Bl6/J mice were orally treated for 14 weeks with either vehicle (n=6) or 50 mg/kg/day of BIO103 (n=4). Immobilized leg: gastrocnemius (A) and plantaris (B) muscles (right rear leg) were isolated and weighed. Active leg: gastrocnemius (C) and quadriceps (D) muscles (left rear leg) were isolated and weighed. Statistical analysis was performed using an unpaired T-test between Adult and Old groups and between Old and Old BIO103 groups. *: *P* <0.05 vs adult; **: *P* <0.01 vs adult

→ Chronic BIO103 oral treatment tends to compensate for the significant atrophy of selected muscles in active and immobilized limbs observed in aging mice.

Plasma measurement

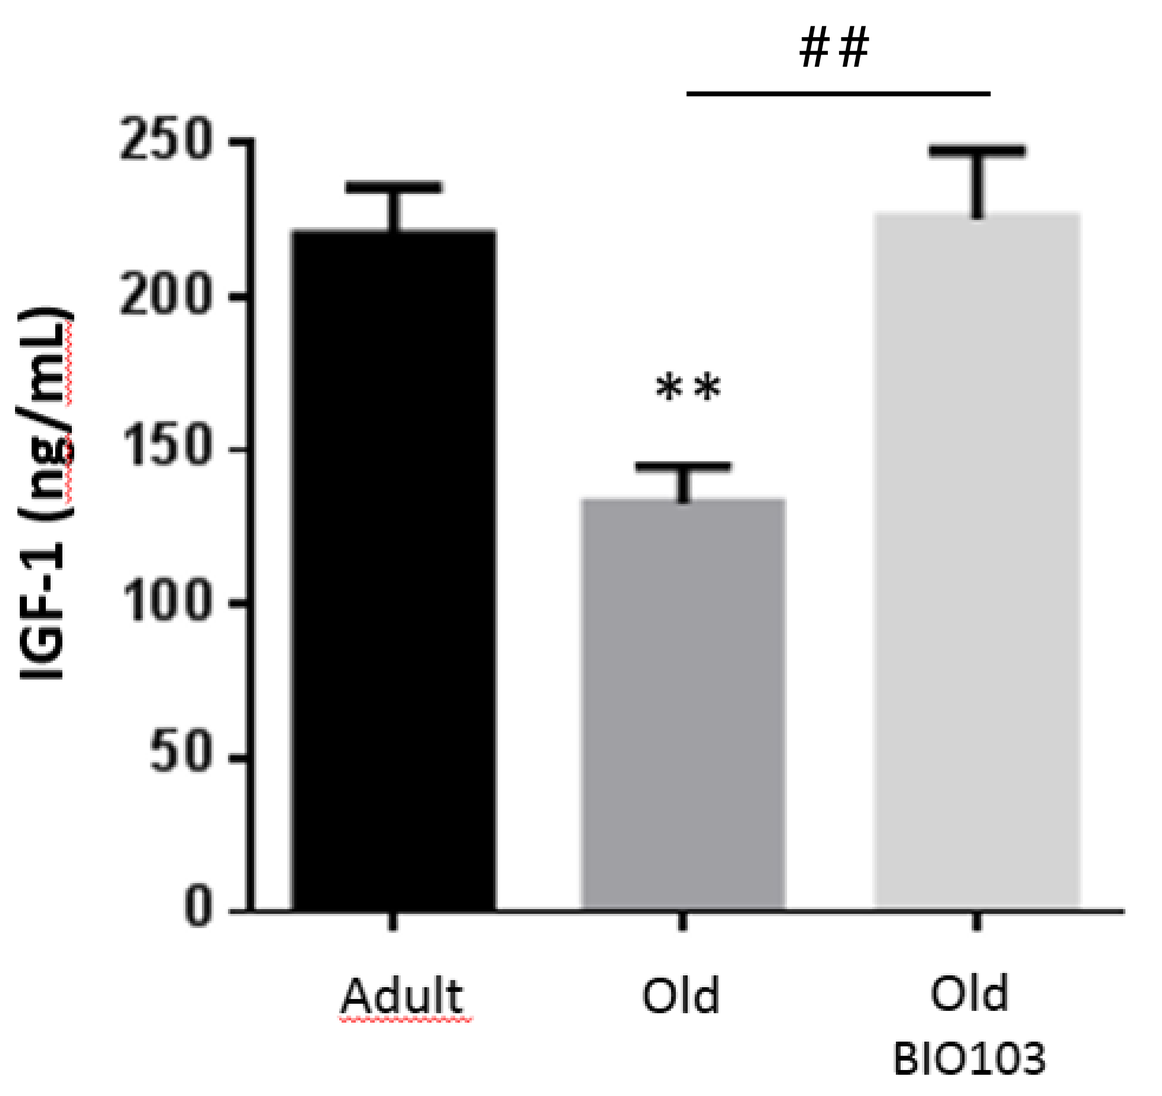
##
IGF-1 (ng/mL)
**
Adult
Old
Old BIO103

Figure 5: Effects of BIO103 on IGF-1 plasma level. Adult (12 months; n=10) and old (22 months) C57Bl6/J mice were orally treated for 14 weeks with either vehicle (n=7) or 50 mg/kg/day of BIO103 (n=7). IGF-1 plasma levels were measured by Elisa assay. Statistical analysis was performed using an unpaired T-test between Adult and Old groups and between Old and Old BIO103 groups. ##p<0.01 vs old. **p<0.01 vs adult

→ In accordance with our data obtained on muscle weight, BIO103 is responsible for a significant increase in animal IGF-1 plasma level in old animals. Importantly, BIO103 compensates for the significant loss of IGF-1 as a consequence of aging.

Functionality test

- *in situ*

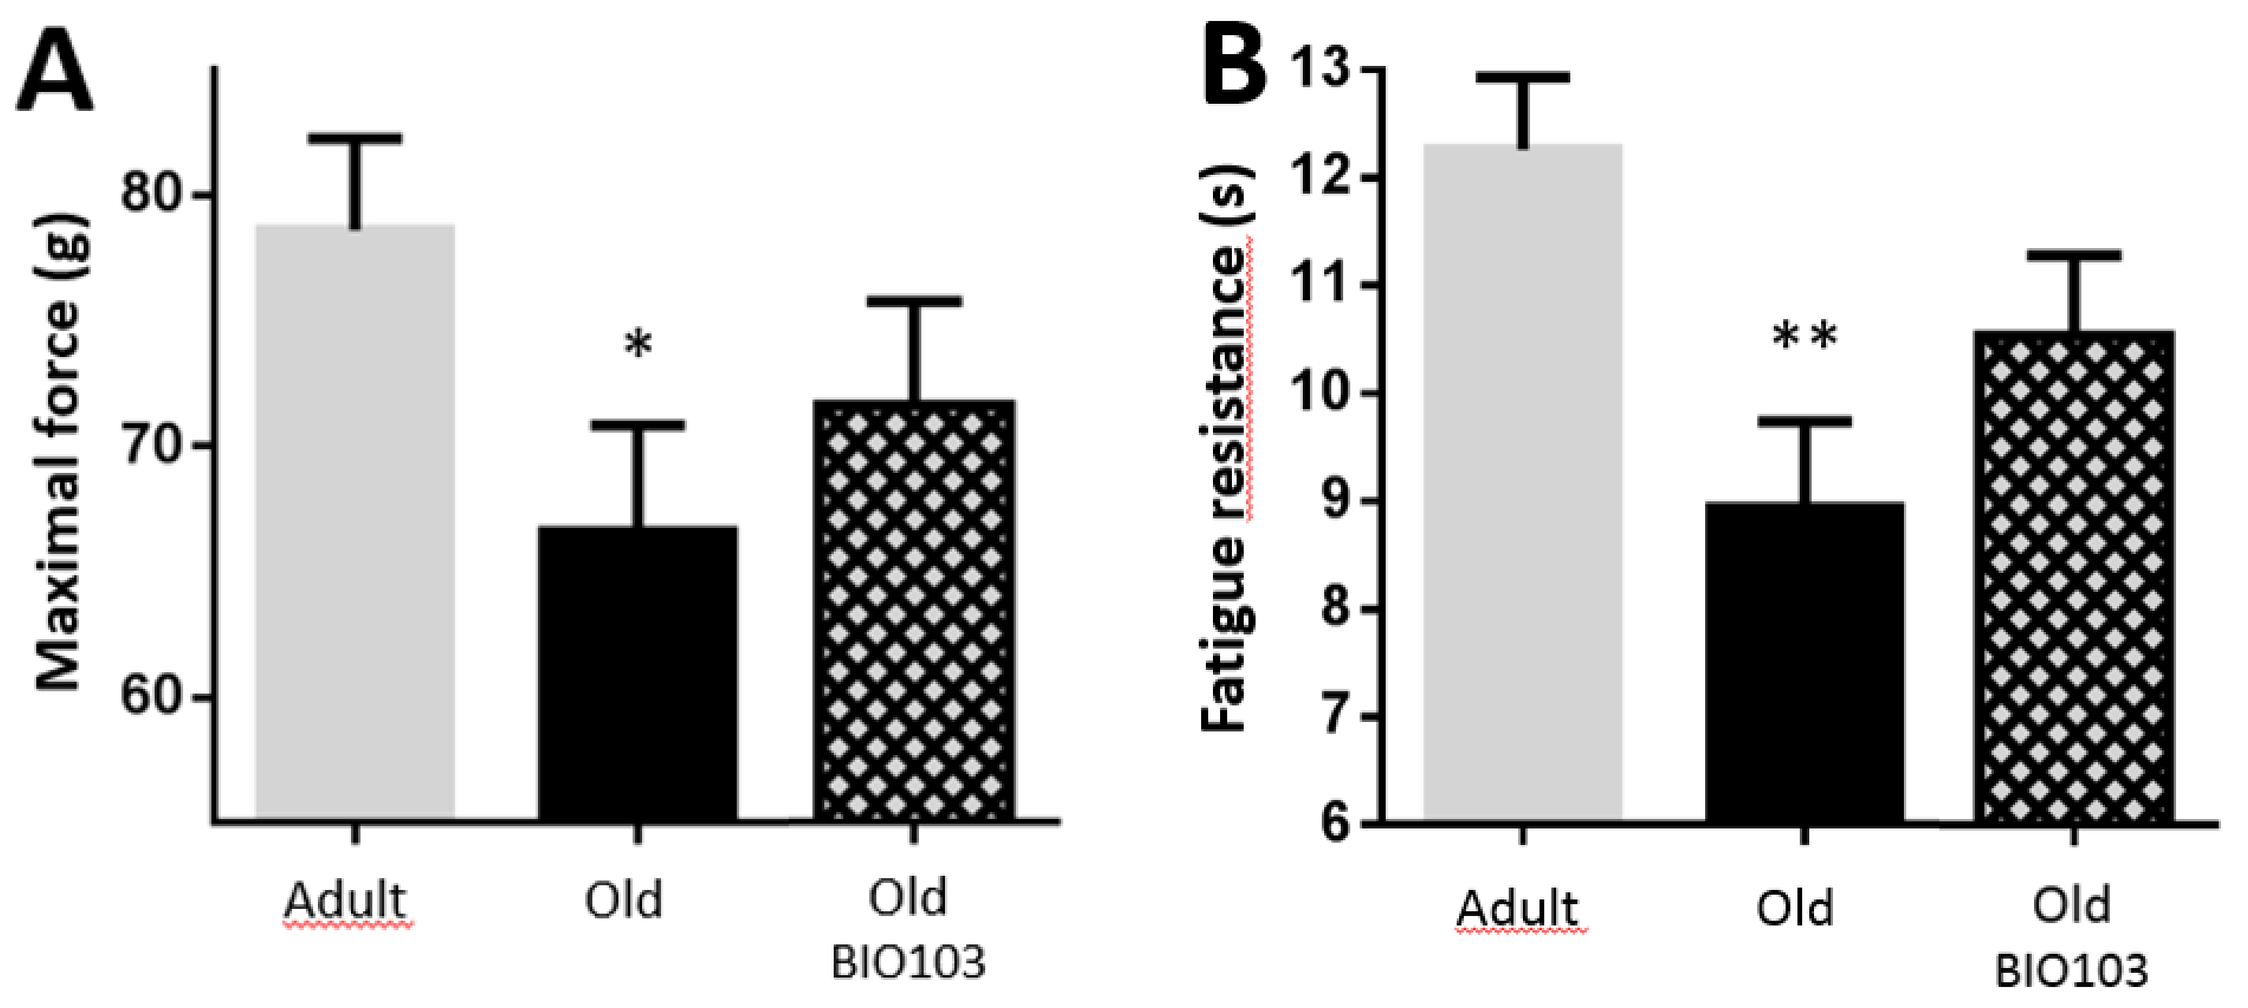
A
Maximal force (g)
*
B
Fatigue resistance (s)
**
Adult
Old
Old BIO103

Figure 6: Effects of BIO103 on *tibialis anterior* muscle maximal force and fatigue resistance in adult and old mice. Adult (12 months; n=10) and old (22 months) C57Bl6/J mice were orally treated for 14 weeks with either vehicle (n=7) or 50 mg/kg/day of BIO103 (n=7). Maximal force (A) and fatigue resistance (B) of *tibialis anterior* muscle from active leg were recorded. Statistical analysis was performed using an unpaired T-test between Adult and Old groups and between Old and Old BIO103 groups. *: *P* <0.05 vs adult; **: *P* <0.01 vs adult.

→ BIO103 treatment tends to compensate for the loss of muscle functionality observed in aging mice.

- *in toto*

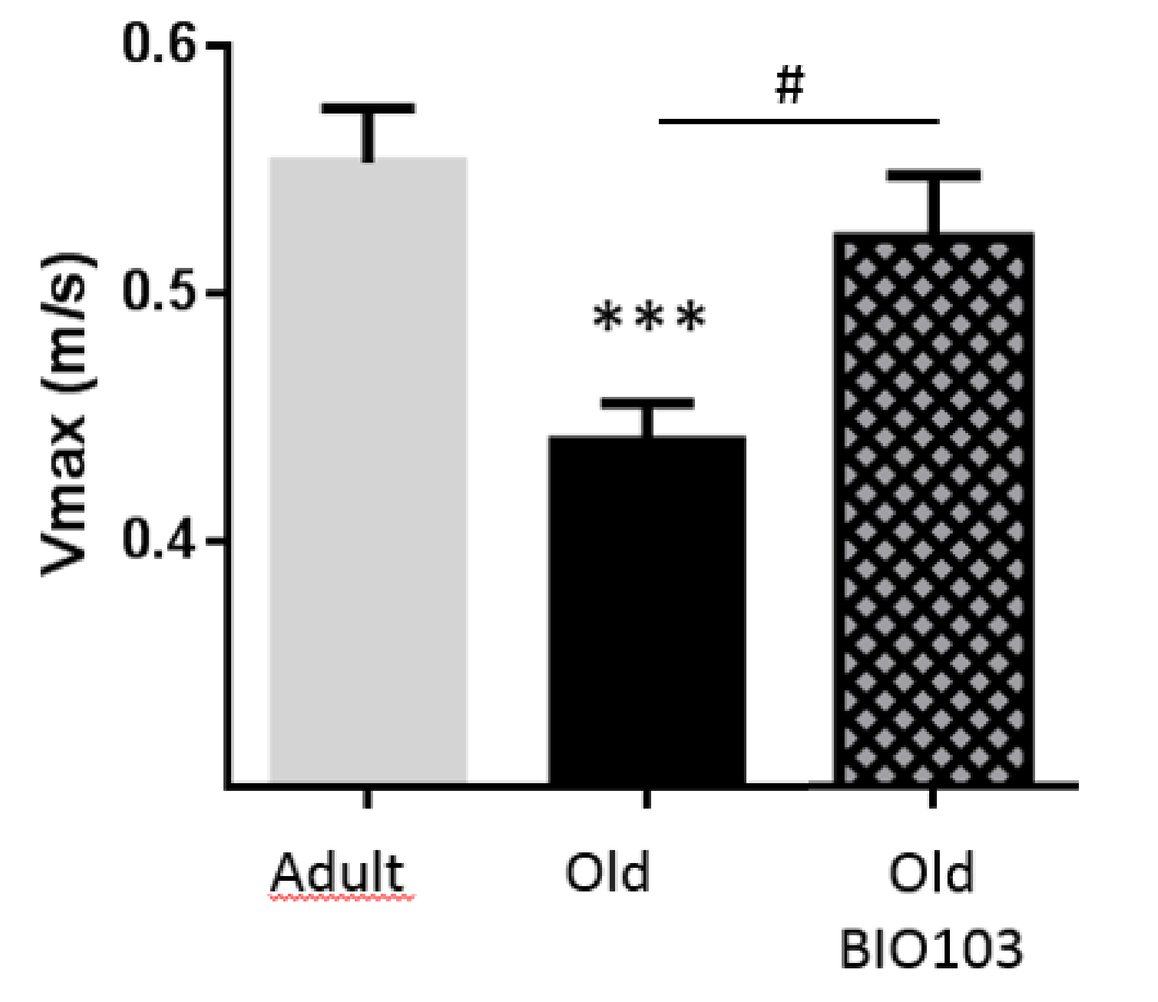
#
Vmax (m/s)

Adult
Old
Old BIO103

Figure 7: Effects of BIO103 on maximal speed in adult and old mice. Adult (12 months; n=10) and old (22 months) C57Bl6/J mice were orally treated for 14 weeks with either vehicle (n=8) or BIO103 (n=8). Maximal speed was recorded on treadmill. Statistical analysis was performed using an unpaired T-test between Adult and Old groups and between Old and Old BIO103 groups. ***: p<0.001 vs adult; #: p<0.05 vs old.

→ BIO103 treatment compensates for the significant loss of running velocity as a consequence of aging.

Conclusion

BIO103, a new orally-available hemi-synthetic derivative of 20-hydroxyecdysone, displayed both *in vitro* and *in vivo* anabolic properties, which translated into improved functional performance in old animals. These investigations demonstrate the potential of BIO103 in improving skeletal muscle quality in aging mammals, and warrant further studies towards its development as a drug candidate for the treatment of sarcopenia.