SARA Clinical program for evaluating safety and efficacy of Sarconeos in a Phase 2b clinical trial

Waly Dioh[1], Carole Margalef[1], Philippe Dupont[1], Pierre Dilda[1], René Lafont[12], Stanislas Veillet[1] and Susanna Del Signore[1]

[1] Biophytis, UMPC – BC9, 4 place Jussieu, 75005 Paris, France. [2] Sorbonne Universités, UPMC Univ Paris 06, Paris-Seine Biology Institut (BIOSIPE), CNRS, 75005 Paris, France

Abstract

Introduction
Sarcopenia is a key underlying cause of physical frailty, a reversible condition in older subjects, which may lead to mobility disability and dependency. Sarcopenia is characterized by the loss of muscle mass and function. Biophytis has developed the drug candidate Sarconeos (BIO101) acting via Mas, the Angiotensin1-7 receptor for prevention and treatment of Sarcopenia. Mas receptor is one of the components of the Renin-Angiotensin System.
Methods
Specifically, the SARA clinical program was developed with Sarconeos (BIO101) in Age-related Sarcopenia, and is composed of three main studies:
The completed SARA-PK Phase 1 study that evaluated Sarconeos safety profile in young and elderly volunteers up to 1400 mg/day in single administration and up to 900 mg/day after 14-day multiple oral administration.
The ongoing SARA-OBS observational study, that aims to validate applicability of inclusion criteria based on sarcopenia definition according to the Foundation of NIH (Studenski et al., 2014) and a very low Short Physical Performance Battery (SPPB; Guralnik J et al., 1994). This study will also evaluate functional (6 minutes walking distance: 6MWD and 400 meters test) and three Patient Reported Outcomes (SF-36; SarQoL, TSD-OC) parameters.
The SARA-INT interventional phase 2 study that will evaluate safety and efficacy of two oral doses of Sarconeos versus placebo in older sarcopenic patients (scheduled for second semester 2017).
All data are hosted within the SARA dedicated platform, allowing real-time monitoring.
Results
Our presentation will focus on 1) Sarconeos effects on a battery of pharmacodynamic plasma biomarkers (Myoglobin, PIIINP etc….) as observed in SARA-PK. 2) Preliminary baseline data of SARA-OBS on target population and primary (6 MWD/400 meters and Patient Reported Outcomes) and secondary endpoints. And, 3) SARA-INT methodology for investigating Sarconeos safety and efficacy in sarcopenic older patients.
Conclusion
The SARA clinical program will allow to outline the effects of Sarconeos on age-related sarcopenia.

Introduction

Characterized by loss of muscle mass and function, sarcopenia is a key underlying cause of physical frailty, a reversible condition in older subjects, which may lead to mobility disability and dependency. Biophytis has developed the drug candidate Sarconeos (BIO101) whose active principle is 20 hydroxyecdysone (20E), purified at 97% from the edible *plant Stemmacantha carthamoides.*

The SARA clinical program, developed for Sarconeos (BIO101) on sarcopenic patients is hosted in the SARA database and is composed of three main studies (see also poster 2-64):

- SARA-INT, the interventional study that evaluates safety and efficacy of Sarconeos in older sarcopenic patients.
- SARA-PK, the Phase 1 study that showed safety and pharmacokinetic profiles of Sarconeos in elderly subjects and that allowed the selection of doses for SARA-INT. Moreover, BIO101 increased plasma levels of (PIIINP) and reduced plasma levels of CK MB and Myoglobin.
- SARA-OBS, the observational study that allows the characterization of population and main parameters of SARA-INT.

Each of SARA-OBS and SARA-INT study is a 6-month multicenter, clinical trial enrolling community-dwelling persons in Europe and USA and aged 65 years and older at risk of mobility disability across. The main objective is to evaluate 2 selected doses of BIO101 in the targeted sarcopenic population.

Methods

SARA-OBS Clinical study design

Sarcopenia is defined according to the criteria of the Foundation of NIH (Studenski et al., 2014); The risk of mobility disability is operationalised using the Short Physical Performance Battery (SPPB; Guralnik et al., 1994)

- SPPB ≤ 8/12
- Absolute ALM (< 19.75 in men and < 15.02 in women) and ALM/BMI (< 0.789 in men, < 0.512 in women) measured by DXA .

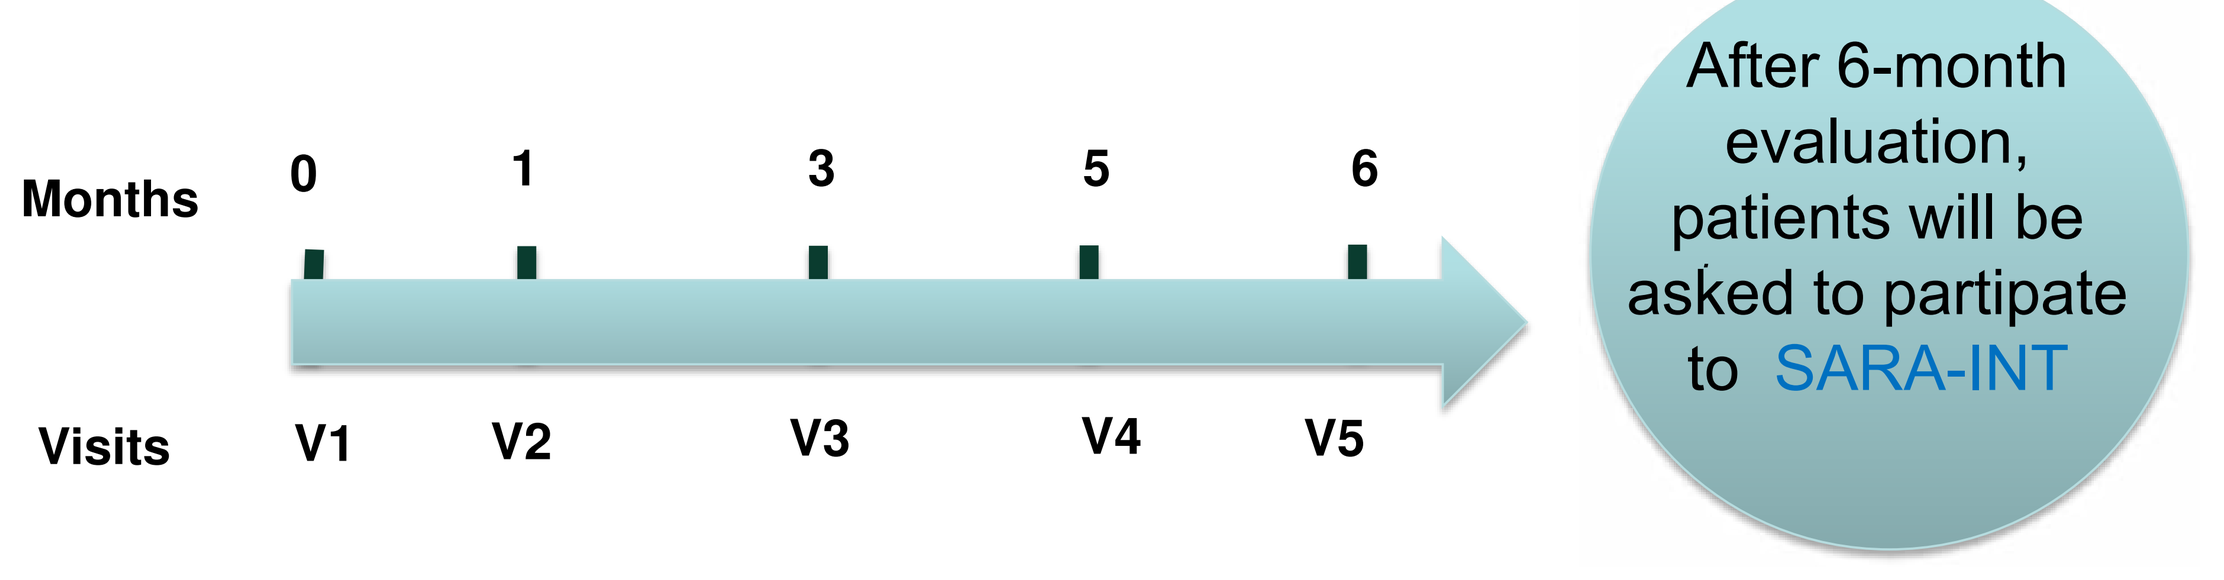
Months
0
1
3
5
6
Visits
V1
V2
V3
V4
V5
After 6-month evaluation, patients will be asked to partipate to SARA-INT

Main objective

- Characterize the target population in Europe and USA.
- Estimate the prevalence of sarcopenia including sarcopenic obesity in a sample of older persons with poor physical function.

Primary endpoint:

- 400 m walking test gait speed
- Patient reported outcomes (PROs): Short Form Health Survey (SF-36) and Sarcopenia Quality of Life (SARQOL) and TSD-OC for BMI ≥ 30

Secondary endpoints

- Body composition, Gait speed; Grip strength, 6 minutes walk

Exploratory Endpoints

- Actimetry; Biomarkers (Myostatin; PIIINP; IL-6; HsCRP; Aldosterone; Renin; Isolated PBMC, etc….)

Results

SARA-OBS Workflow

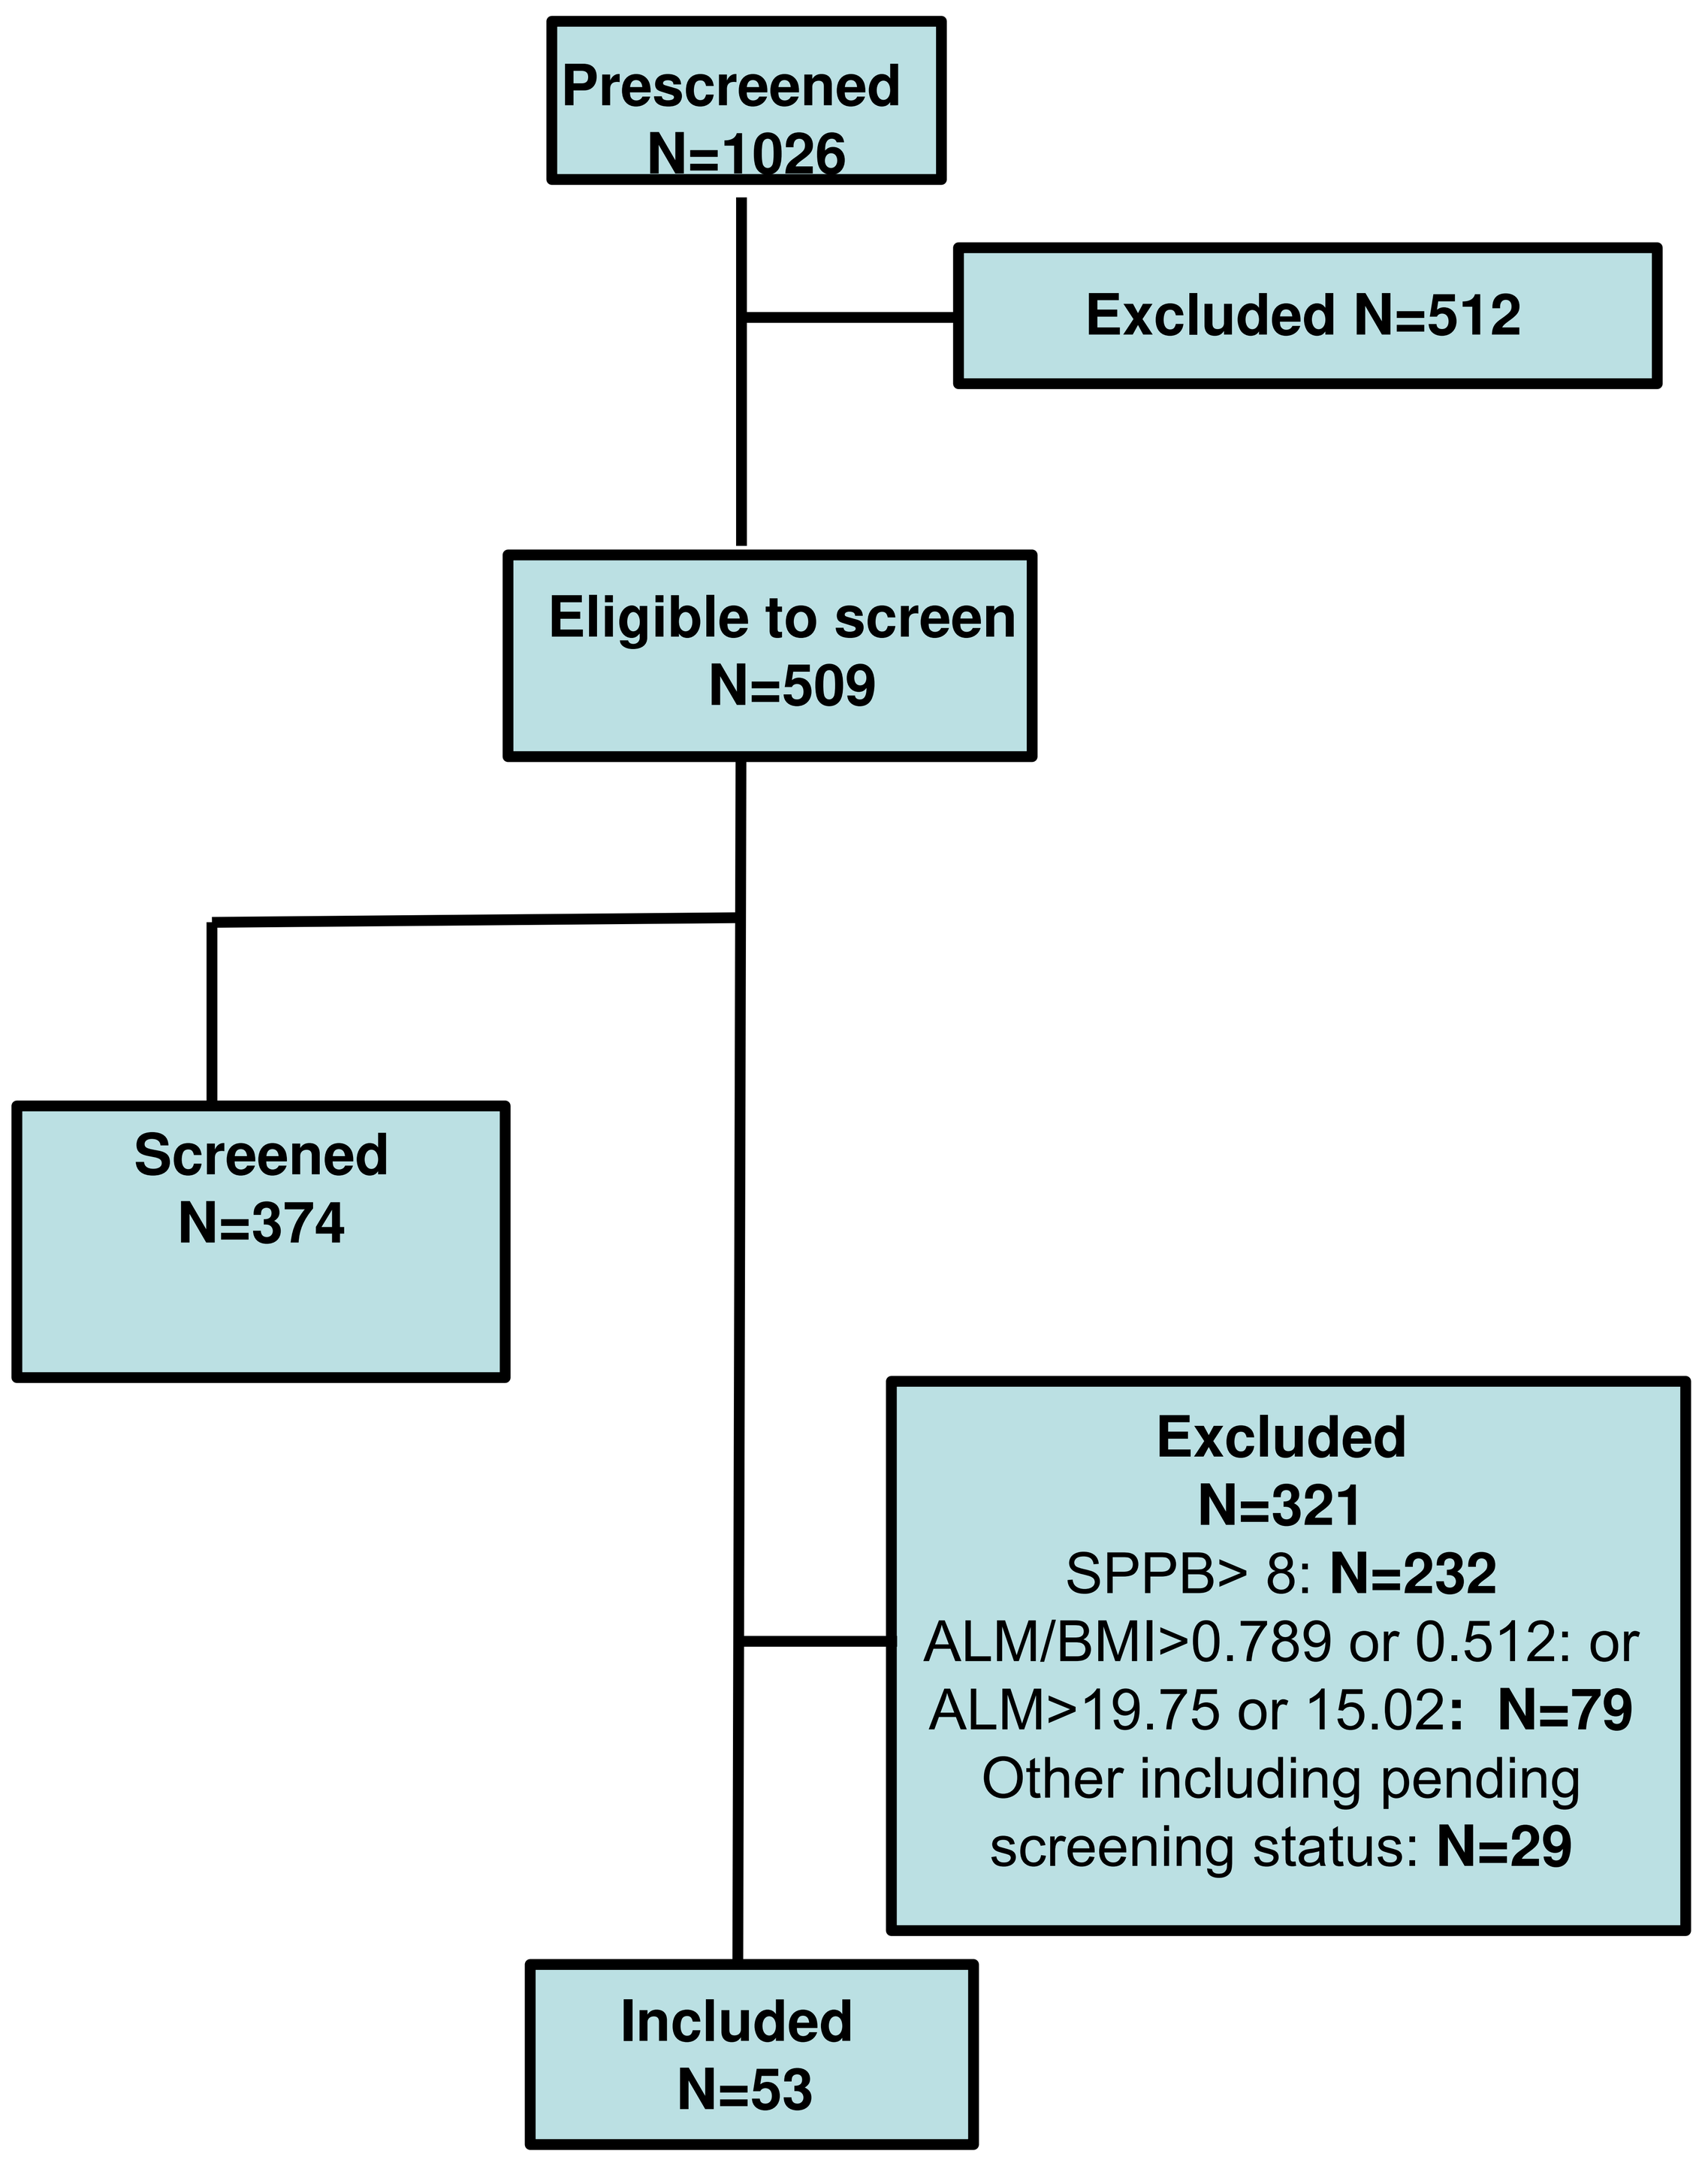
Prescreened
N=1026
Excluded N=512
Eligible to screen
N=509
Screened
N=374
Excluded
N=321
SPPB> 8: N=232
ALM/BMI>0.789 or 0.512: or
ALM>19.75 or 15.02: N=79
Other including pending
screening status: N=29
Included
N=53

- 61% of the prescreened patients were not retained. Main reasons were disease of the exclusion criteria, the absence of mobility issues.
- Prescreening failure (Prescreened vs included) was rather high: only 49% of prescreened were selected.
- Screening failure was high: only 15 % of screened patients were included. This rate was also observed in other sarcopenia clinical trials (Fielding et al., 2015; Fielding et al., 2017).
- Main reasons of screening failure were SPPB>8 (61%); ALM/BMI > 0.789 or > 0.512 and ALM > 19.75 kg or > 15.02 kg (24%) and Other : 30%

Baseline characteristics of SARA-OBS included patients

After an average period of 4 months post SIV, the following characteristics can be outlined

Characteristics	Absolute	Standard Deviation
Age	78.65	7.86
BMI	30.78	7.49
Male : Female	23:29	NA
SPPB	6.57	1.72
Gait Speed in SPPB (sec)	0.75	0.19
Chair stand SPPB	1.68	0.82
Men	1.79	0.85
Women	1.59	0.80
Appendicular Lean Mass (ALM)	16.59	4.96
Men	19.26	5.33
Women	14.37	3.33
ALM/BMI	0.57	0.15
Men	0.66	0.15
Women	0.49	0.09
6MWD	314.86	103.19
400M (min)	7.88	3.61
Gait speed 400M (m/sec)	0.88	0.27

- As seen in other large sarcopenia clinical trials, patients showed high BMI.
- 400 m gait speed was equivalent to the Life study (0.83 m/s at baseline Pahor et al., 2014)
- Mean SPPB is rather low (6.57/12), corresponding to patients at risk of mobility disability and comparable to other sarcopenia studies (the Life study with 7.4±1.6 in the physical activity group, Pahor et al., 2014).
- Gait speed SPPB is <0.8 m/s, fitting with the definition of sarcopenia from the EWGSOP.
- ALM/BMI in men was lower than the FNIH threshold (0.66 vs 0.789) and similar in women (0.49 vs 0.52)
- The 6MWD at 314.86 meters was expected for patients of the mean age (78.65 years) and BMI (30.78)

Next Steps

SARA-INT clinical study design

- SARA-INT is an EU and US double-blind, placebo controlled, randomized INTerventional Clinical trial.
- SARA-INT aims to evaluate safety and efficacy of Sarconeos (BIO101), the investigational drug orally administered for 6 month.
- SARA-INT is involving at least 21 sites.
- Most of the sites are currently recruiting in SARA-OBS, and Additional sites are currently being selected

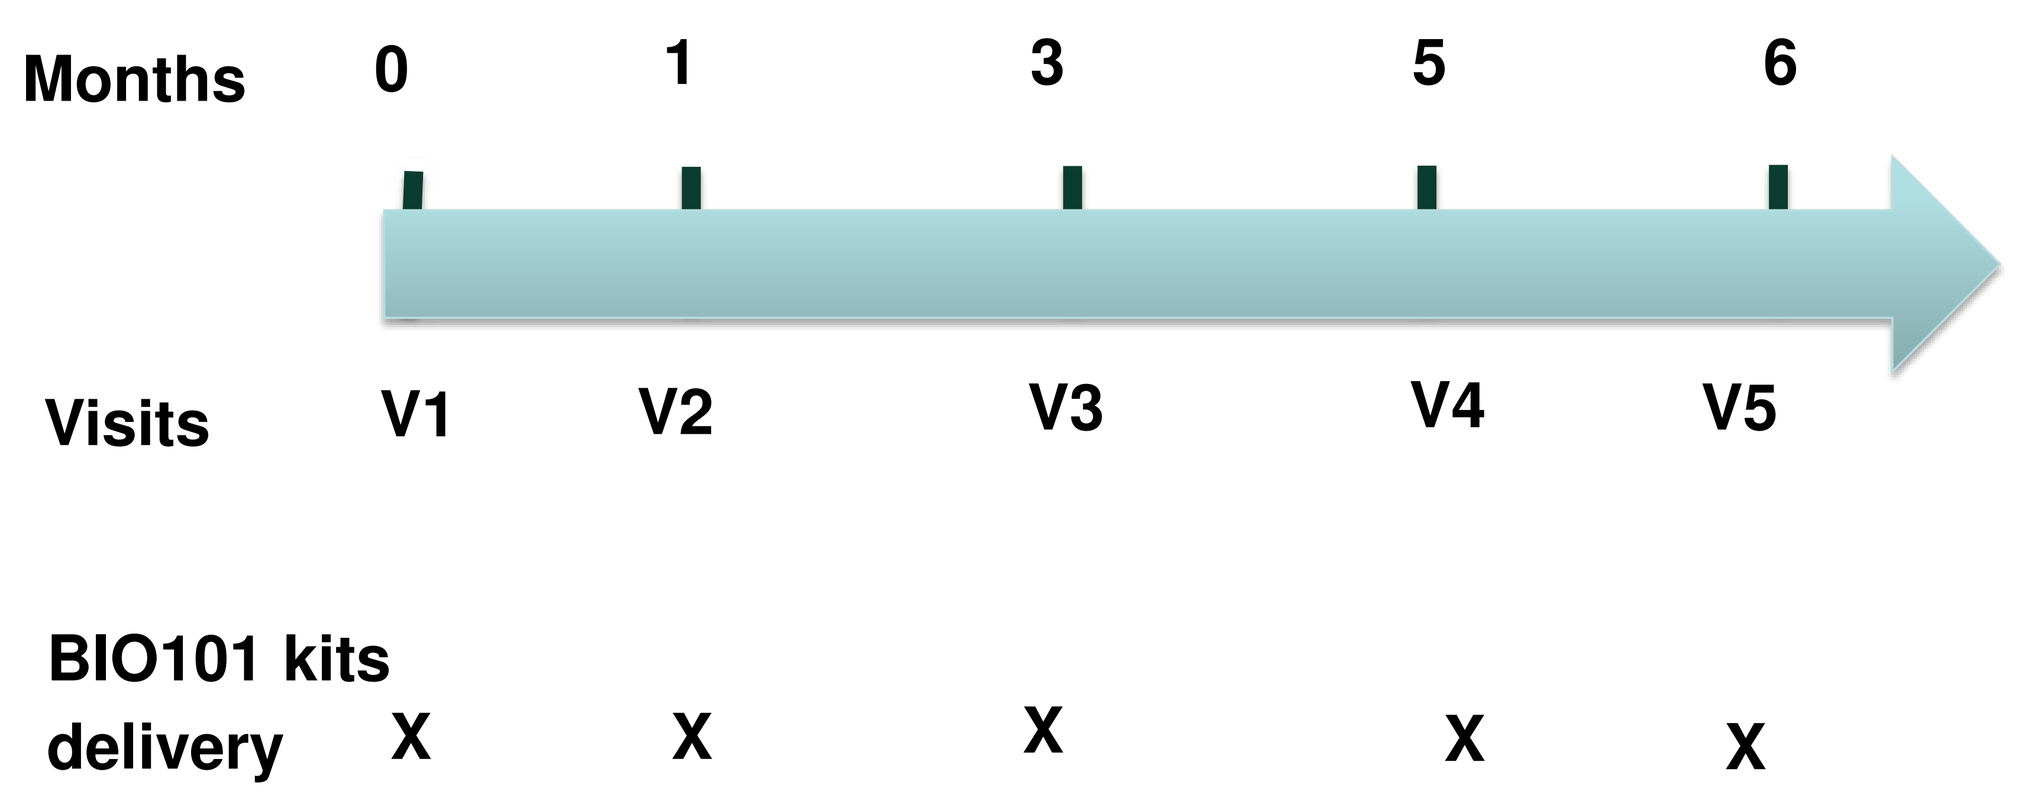
Months
0
1
3
5
6
Visits
V1
V2
V3
V4
V5
BIO101 kits delivery
X
X
X
X
X

Primary objectives:

- Evaluate the effect of two daily doses of BIO101 versus placebo on gait speed at the 400MW test.

Key secondary objectives

- Compare the change from baseline of the Patient Reported Outcome (PRO): The Physical Function Domain (**PF-10**) of the Short Form Health Survey (SF-36).
- Rising from a chair

Other Secondary , Tertiary and Exploratory Objectives

Body composition; Muscle strength; stair climbing; SPPB; PRO (SF-36, SarQoL, TSD-OC); Actimetry; Biomarkers (Myostatin; PIIINP; IL-6; HsCRP; Aldosterone; Renin; Isolated PBMC, etc….)

Target population

334 sarcopenic patients reporting loss of physical function and considered at risk of mobility disability will be recruited for the SARA-INT study.

The recruitment criteria are similar to the Foundation of NIH (Studenski et al., 2014)

- SPPB ≤ 8/12
- Absolute ALM (< 19.75 in men and < 15.02 in women)
- and ALM/BMI (< 0.789 in men, < 0.512 in women) measured by DXA

Conclusions

- Preliminary observations in SARA-OBS confirmed recruitment opportunities in sarcopenia trials when using the FNIH criteria (Studenski et al., 2014, ALM/BMI<0.789 in Men and <0.512 in Women) and Absolute ALM <19.75 kg in men and <15.02 kg in women.
- Similarly to SPRINTT study, we accepted the challenge to select low performers at SPPB<8/12 as an index of mobility disability risk.
- Baseline characteristics are comparable to other sarcopenia studies (Life study, Pahor et al., 2014).
- Target population derived from SARA-OBS will be eligible to investigate the drug candidate Sarconeos (BIO101) in the SARA-INT phase 2 interventional study.

• References

- Fielding RA, Travison TG, Kirn DR et al. Effect of Structured Physical Activity and Nutritional Supplementation on physical Function in Mobility-Limited Older ADULTS: results from the VIVE2 Randomized trial. J Nutr Healthy Aging. 2017. Online First Articles. 1-7
- Fielding RA, Guralnik JM, King AC, Pahor M, McDermott M, Tudor-Locke C, Manini TM, Glynn NW, Marsh PA, Axtell RS, Hsu Fang-Chi, Rejeski WJ, For the LIFE study group. Dose of physical activity, physical functioning and disability risk in mobility-limited older adults: Results from the LIFE study randomized trial PLOS One August 2017
- Studenski SA, Peters KW, Alley DE, Cawthon PM, McLean RR, Harris TB, Ferrucci L, Guralnik JM, Maren, Fragala, MS, Kenny AM, Kiel, DP, Kritchevsky SB, Shardell MD, Dam T-T L, and Vassileva MT. The FNIH Sarcopenia Project: Rationale, Study Description, Conference Recommendations, and Final Estimates J Gerontol A Biol Sci Med Sci. 2014; 69(5): 547-558
- Guralnik JM, Simonsick EM, Ferrucci L, Glynn RJ, Berkman LF, Blazer DG, Scherr PA, Wallace RB. A short physical performance battery assessing lower extremity function: association with self-reported disability and prediction of mortality and nursing home admission. J Gerontol. 1994 Mar;49(2):M85-94. PubMed PMID: 8126356.
- Pahor M, Guralnik JM, Ambrosius WT, Blair S, Bonds DE, Church TS, Espeland MA, Fielding RA, Gill TM, Groessl EJ, King AC, Kritchevsky SB, Manini TM, McDermott MM, Miller ME, Newman AB, Rejeski WJ, Sink KM, Williamson JD; LIFE study investigators. Effect of structured physical activity on prevention of major mobility disability in older adults: the LIFE study randomized clinical trial. JAMA. 2014 Jun 18;311(23):2387-96